ID:1553170

The Economic Benefits of Time Off Treatment: Real-world HRU, Costs, and Subsequent Treatment with Fixed Duration Venetoclax among Patients with Chronic Lymphocytic Leukemia

Alan P. Skarbnik[1]*, Lori A. Leslie[2], Nnadozie Emechebe[3], Dureshahwar Jawaid[3], Beenish S. Manzoor[3], Hasan Alhasani[3], Brian T. Hill[4]

[1]Novant Health, Charlotte, NC, USA; [2]John Theurer Cancer Center, Hackensack University Medical Center, Hackensack, NJ, USA; [3]AbbVie, North Chicago, IL, USA; [4]Cleveland Clinic, Cleveland, OH, USA

*Presenting author

OBJECTIVE

To quantify the healthcare resource utilization and costs during and post-completion of venetoclax plus obinutuzumab (VO) or venetoclax plus rituximab (VR) therapy and evaluate subsequent treatment patterns in patients with chronic lymphocytic leukemia (CLL)/small lymphocytic leukemia (SLL) in first-line and relapsed/refractory settings over an extended period

CONCLUSIONS

Being off treatment with fixed treatment duration (FTD) therapy has multiple benefits, such as economic, clinical, and humanistic, and this real-world study is one of the first to illustrate the economic benefits of time off-treatment as afforded by an FTD regimen

In this updated real-world cohort, healthcare costs were reduced when patients completed or were off treatment

Patients treated with VO and VR had low rates of subsequent therapy during the available follow-up, consistent with previous findings, suggesting these are effective therapies for patients with CLL

Compared to indefinite therapy with Bruton's tyrosine kinase–inhibitor treatment, these results may carry implications not only for payers and access decision-makers, but also considerations for shared decision-making between physicians and patients

If you have any questions/comments regarding this presentation, please contact Dr Alan P. Skarbnik on azskarbnik@novanthealth.org

For additional information or to obtain a PDF of this poster

Scan QR code or utilize the following link to download an electronic version of this presentation and another AbbVie iWCLL 2023 scientific presentation:

https://abbvie1.outsystemsenterprise.com/GMAEvent Publications/Assets.aspx?ConferenceId=652

Copies of this poster obtained through the QR code are for personal use and may not be reproduced without permission from iwCLL and the author of this poster

QR code expiration: September 06, 2024

To submit a medical question, please visit www.abbviemedinfo.com

Presented at the International Workshop on Chronic Lymphocytic Leukemia (iwCLL), October 7-9, 2023, Boston, Massachusetts, USA

Venetoclax is developed in a collaboration between AbbVie Inc. and Genentech Inc. AbbVie funded the study and participated in the study design, research, analysis, data collection, interpretation of the data, reviewing, and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship.

Medical writing support was provided by Rabiah Bhandari, MSc, of Fishawack Facilitate Ltd, part of Fishawack Health, and funded by AbbVie Inc.

APS: Research support: AbbVie, Pharmacyclics, Verastem, Acerta Pharma, AstraZeneca, Celgene, Kite Pharma, and Bristol Myers Squibb; Honoraria: AbbVie, Pharmacyclics, Genentech, Seattle Genetics, Novartis, Verastem, Jazz Pharmaceuticals, AstraZeneca, Celgene, Kite Pharma, BeiGene, Gilead Sciences; Consultancy: AbbVie, Pharmacyclics, AstraZeneca, Kite Pharma, Celgene, Genentech, Jazz Pharmaceuticals, Alexion; Speaker and consulting fees: Lilly, ADC Therapeutics. **LAL**: Speaker Bureau: Seattle Genetics, Celgene/BMS, KitePharma, BeiGene, Pharmacyclics/Janssen, AstraZeneca, Epizyme, Karyopharm; Advisory Board and/or consultancy: Bayer, Seattle Genetics, ADC herapeutics, AbbVie, Janssen, Pharmacyclics, Kite, AstraZeneca, TG Therapeutics. **NE, DJ, BSM, HA**: employees of AbbVie and may hold AbbVie stock and/or stock options. **BTH**: Research funding and consulting fees from AbbVie, Genentech, Pharmacyclics, AstraZeneca, and BeiGene.

References

1. Davids MS, Manzoor BS, Hazra NC, et al. *J Clin Pathways* 2022;8(5):36–46.
2. Cho SK, Manzoor BS, Sail KR, et al. *Pharmacoeconomics* 2020;38(9):941–951.
3. Allan JN, Skarbnik AP, Emechebe N, et al. *Hemasphere* 2022;6:2222–2223.

INTRODUCTION

- Fixed-duration (FD) venetoclax-based regimens are approved for the treatment of chronic lymphocytic leukemia/small lymphocytic leukemia (CLL/SLL) in first-line (1L) as well as relapsed/refractory (R/R) settings
- Prior studies have described FD treatment as related to clinical trials or total cost of care
 - For example, the 10-year cumulative costs of treatment sequences starting with fixed treatment duration (FTD) venetoclax plus obinutuzumab (VO) resulted in lower costs compared with other novel agents[1] and introduction of FTD VO for 1L CLL treatment resulted in substantial cost savings over a 3-year time horizon[2]
- However, there are limited data characterizing the real-world benefits of FTD with venetoclax regimens
- Preliminary data showing reduced healthcare resource utilization (HRU) and cost burden with venetoclax-based regimens suggest that FTD of venetoclax regimens may provide off-treatment economic benefits and this analysis adds to the growing body of evidence on the holistic benefit of time off treatment[3]

METHODS

Study design and data source

- Retrospective observational study using Optum Clinformatics® Data Mart data to identify patients with confirmed CLL/SLL between 1/1/2010 – 6/30/2021 who initiated VO in 1L or venetoclax plus rituximab (VR) in R/R settings
 - Confirmed CLL was defined as two claims at least 30 days apart or one inpatient admission with an ICD-10 code for CLL (C91.1x) or SLL (C83.0x)
- This analysis is an update of a previous cohort with an extended identification period allowing for the identification of 115 additional patients (total N=223)

Inclusion Criteria
- ≥18 years of age
- ≥1 inpatient or ≥2 outpatient claims ≥30 days apart and within 12 months of the first claim with a CLL/SLL diagnosis code
- ≥1 claim with CLL diagnosis post-initiation of the line of therapy for R/R CLL cohort

Exclusion Criteria
- Evidence of acute myeloid leukemia [ICD10:C92.*; ICD9:205.0*]
- Prior treatment for CLL/SLL for 1L cohort only
- Clinical trial participation

- Patients were continuously enrolled in their health plan for 1 year pre- and post-initiation with VO or VR

Follow-up period
- The follow-up period was stratified into two phases:
 - On-treatment phase, defined as time in months from venetoclax initiation to end of treatment
 - Off-treatment phase, defined as time in months from 1 day after the end of venetoclax treatment to end of continuous enrollment/1 day before next line of therapy, or death

Study outcomes

- Mean per-patient, per-month (PPPM) all-cause HRU and costs in both on- and off-treatment phases
 - Total cost was inclusive of medication costs and medical costs, and medical costs included costs incurred during inpatient, outpatient, and emergency room visits
- Subsequent treatment patterns following initiation with VO or VR

Data analysis

- Data were summarized using mean, standard deviation, or median, and interquartile range (IQR)
- The proportion of patients who received subsequent lines of therapy after initiation with VO or VR were reported

RESULTS

Study population

- Baseline data are shown in **Table 1**

VO cohort
- In 1L, 115 VO-treated patients with a median follow up of 23.3 months were identified, with a median age at venetoclax initiation of 70 years, and 71.3% were male
- The median (IQR) duration of the on- and off-treatment phases were 12.4 (11.3–13.5) and 10.2 (5.6–15.1) months, respectively

VR cohort
- In the R/R setting, 108 patients with a median follow-up of 24.0 months, median age at VR initiation of 73 years, and 58.3% male were identified
- The median (IQR) duration of the on- and off-treatment phases were 16.1 (11.0–23.6) and 0.2 (0.0–9.9) months

Table 1. Baseline demographics and clinical characteristics for patients in the VO and VR cohorts

Characteristic	1L VO cohort (N=115)	R/R VR cohort (N=108)
Median (IQR) age at Ven initiation, years	70 (66–74)	73 (65–77)
Sex, n (%)		
Female	33 (28.7)	45 (41.7%)
Male	82 (71.3)	63 (58.3%)
Clinical history		
Renal impairment pre Ven, n (%)	23 (20)	36 (33.3%)
Cardiac comorbidity pre Ven, n (%)	53 (46.1)	61 (56.5%)
CCI without primary malignancies, median (IQR)	1 (0-2)	1 (0-3)
Year of Ven initiation, n (%)		
2016	-	2 (1.9%)
2017	1 (0.9)	-
2018	-	17 (15.7%)
2019	21 (18.3)	31 (28.7%)
2020	60 (52.2)	31 (28.7%)
2021	33 (28.7)	27 (25.0%)
Time to Ven initiation in months, median (IQR)	15.3 (2.4-35.7)	39 (21.8-62.4)
Time on Ven treatment in months, median (IQR)	12.4 (11.3-13.5)	16.1 (11.0-23.6)
Time off Ven treatment in months, median (IQR)	10.2 (5.6-15.1)	0.23 (0-9.9)
Follow-up in months, median (IQR)	23.3 (19.6-28.1)	24.0 (17.8-34.7)

1L, first-line; CCI, Charleston Comorbidity Index; IQR, interquartile range; R/R, relapsed/refractory; VO, venetoclax plus obinutuzumab; VR, venetoclax plus rituximab; Ven, venetoclax

IL VO COHORT

PPPM all-cause costs

- Mean PPPM all-cause costs were $17,186 and $4,927 in the on- and off-treatment phases, respectively, representing a 71% reduction in costs after treatment completion (**Figure 1**)
- The on-treatment costs were driven mainly by medication costs ($14,230 PPPM) relative to medical costs ($2,956 PPPM), while medical costs ($4,106 PPPM) accounted for most of the off-treatment costs (**Figure 1**).

Figure 1. Mean PPPM all-cause costs ($) between the on- versus off-treatment phases for patients in the 1L setting

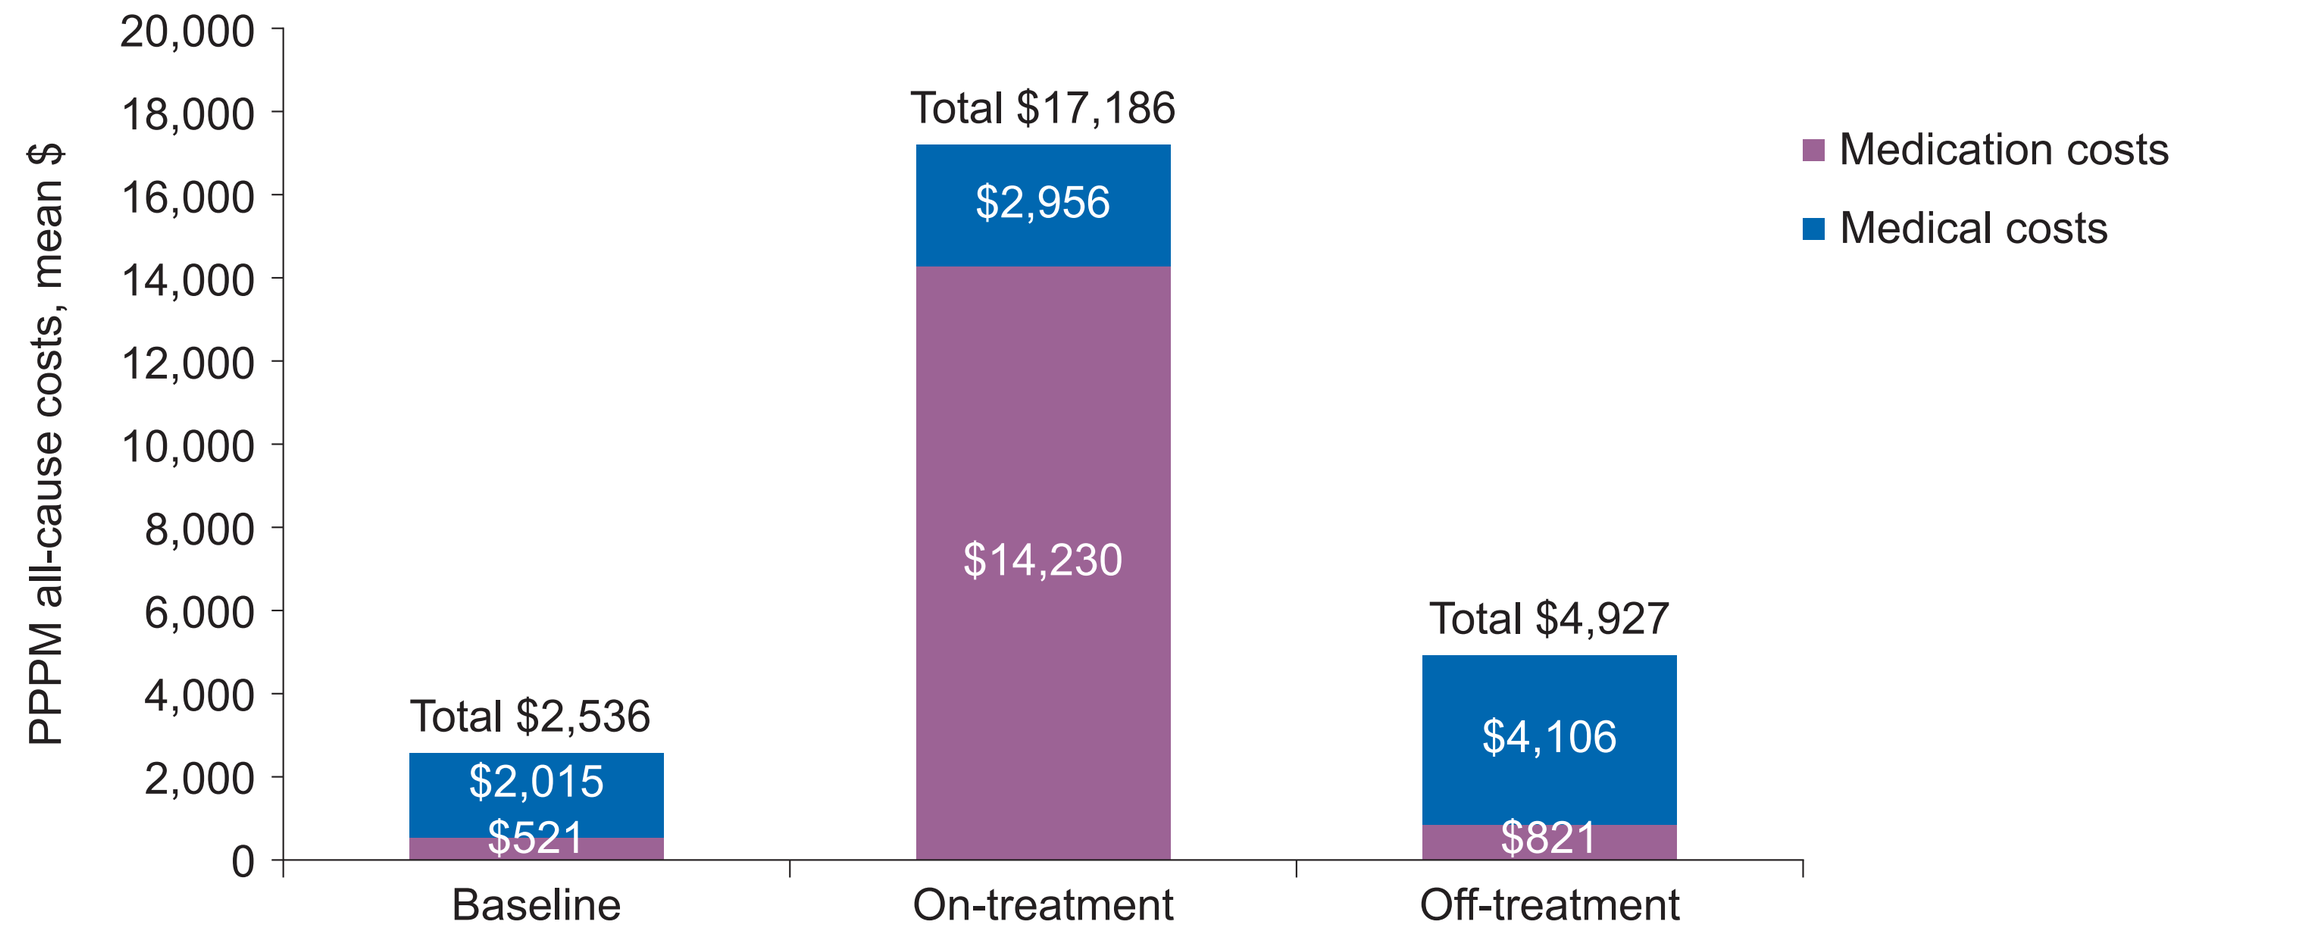

PPPM all-cause costs, mean $
20,000
18,000
16,000
14,000
12,000
10,000
8,000
6,000
4,000
2,000
0
Total $2,536
$2,015
$521
Total $17,186
$2,956
$14,230
Total $4,927
$4,106
$821
Baseline
On-treatment
Off-treatment
Medication costs
Medical costs

Individual components contributing to medical costs, mean (SD)

Medical costs	Baseline[a]	On-treatment	Off-treatment
Inpatient	$593 (1,830)	$791 (2,333)	$2,744 (10,599)
Outpatient	$1,154 (926)	$1,918 (1,458)	$1,028 (1,735)
Emergency room	$111 (317)	$121 (344)	$116 (271)

[a]Baseline entails the 12 months prior to VO initiation
1L, first-line; PPPM, per patient per month; SD, standard deviation

PPPM all-cause HRU

- The mean PPPM all-cause outpatient visits were higher in the on- versus off-treatment phases (3.3 vs 1.8 visits)

Subsequent therapies

- In patients treated with 1L VO, only 9/114 (7.9%) patients moved on to a second line of therapy (**Figure 2**)
 - Only 2 of these 9 patients received 3L therapy (zanubrutinib or VO)

Figure 2. Treatment sequencing for patients who received 1L VO

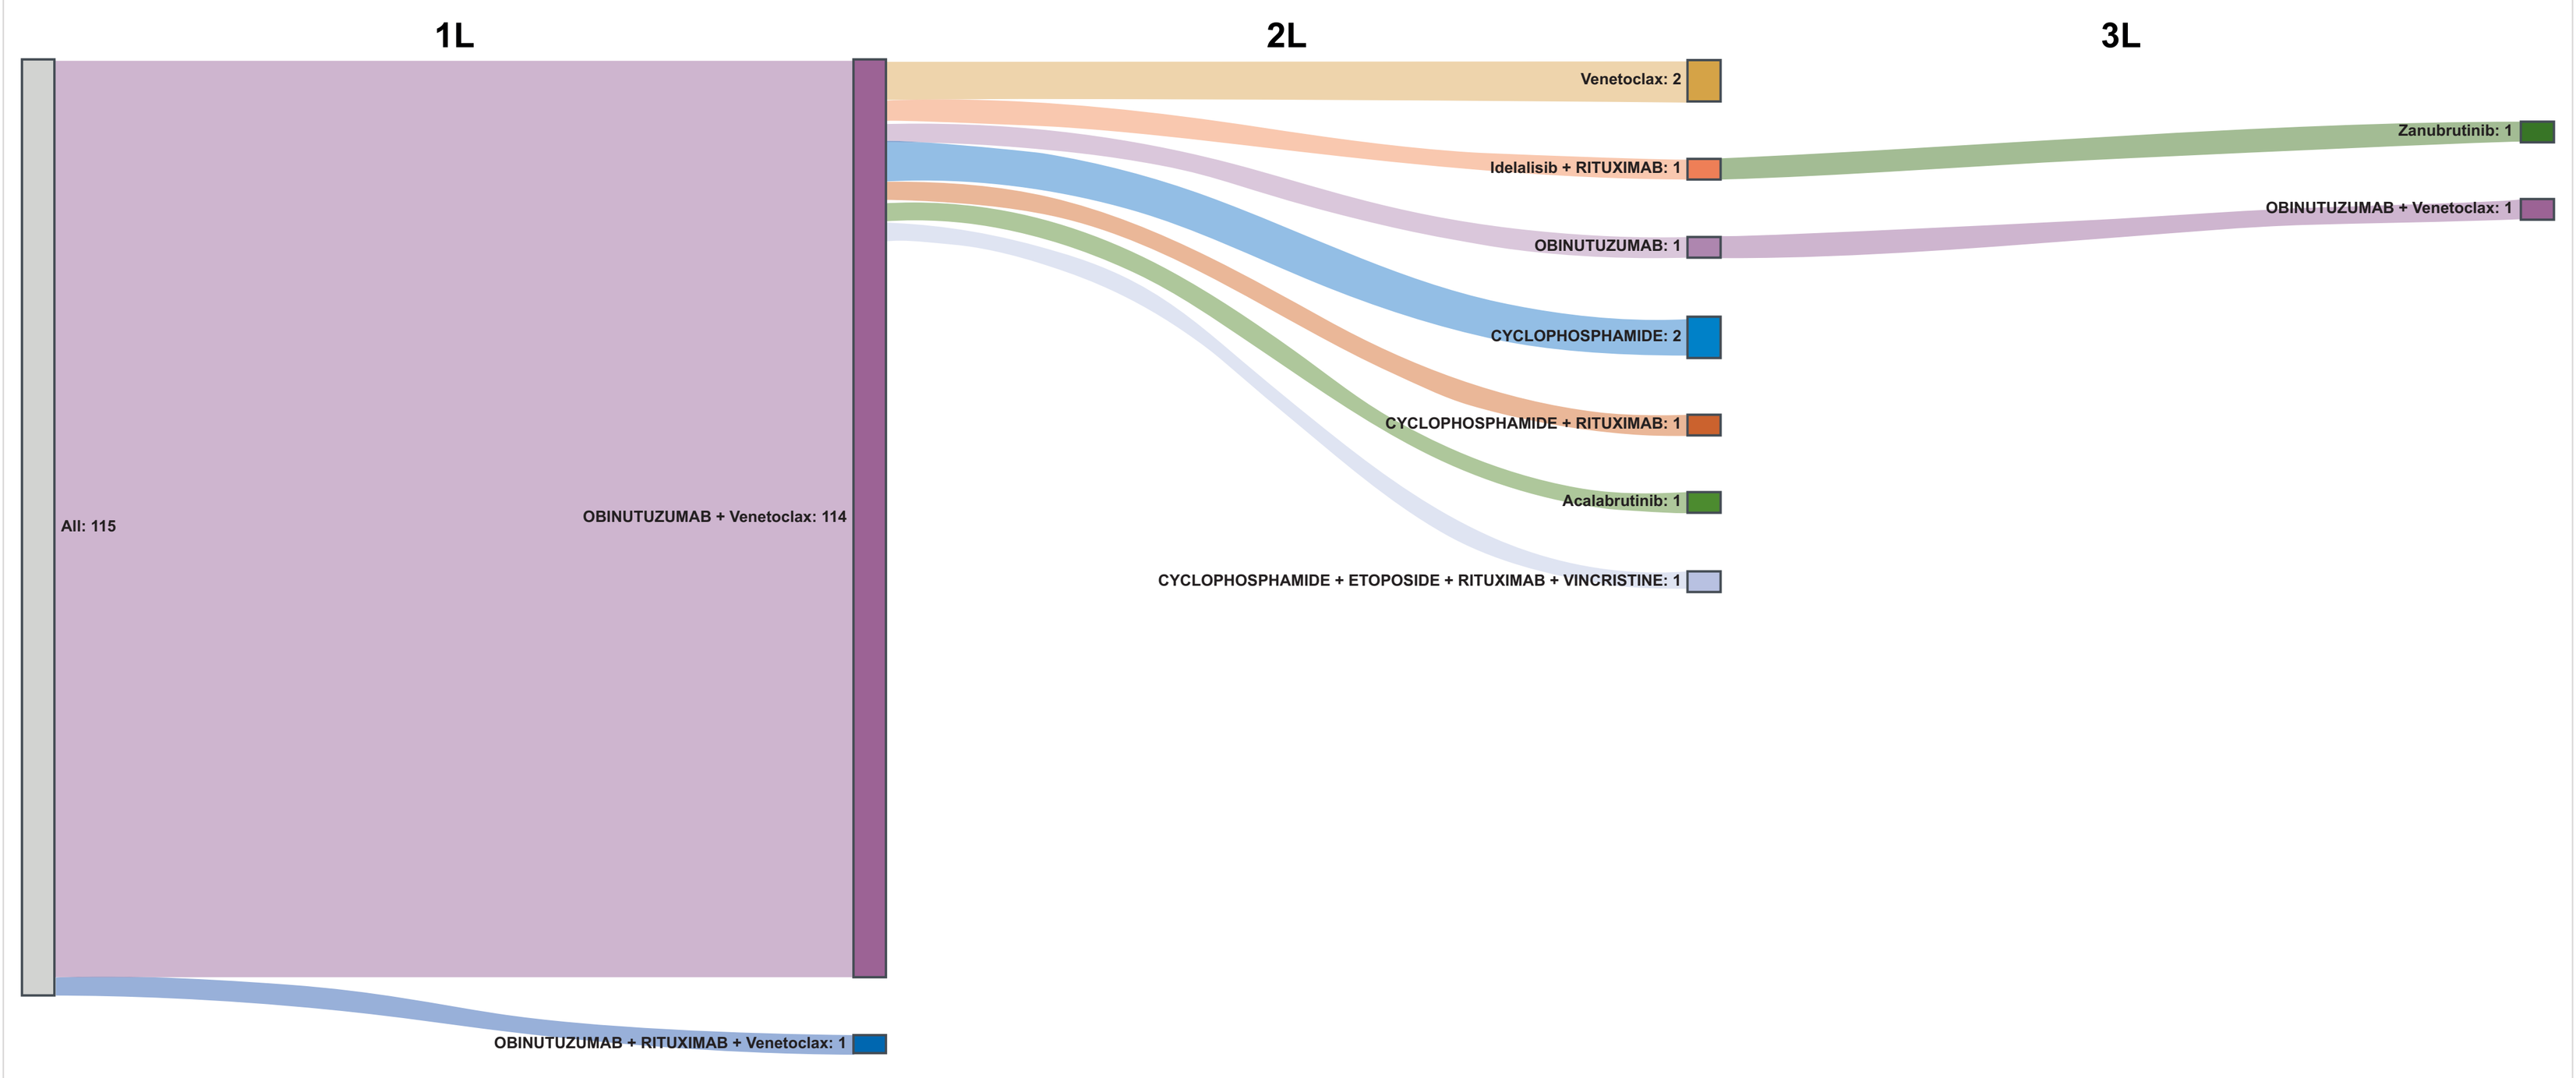

1L
2L
3L
All: 115
OBINUTUZUMAB + Venetoclax: 114
OBINUTUZUMAB + RITUXIMAB + Venetoclax: 1
Venetoclax: 2
Idelalisib + RITUXIMAB: 1
OBINUTUZUMAB: 1
CYCLOPHOSPHAMIDE: 2
CYCLOPHOSPHAMIDE + RITUXIMAB: 1
Acalabrutinib: 1
CYCLOPHOSPHAMIDE + ETOPOSIDE + RITUXIMAB + VINCRISTINE: 1
Zanubrutinib: 1
OBINUTUZUMAB + Venetoclax: 1

1L, first-line; 2L, second-line; 3L, third-line; VO, venetoclax plus obinutuzumab

R/R VR COHORT

PPPM all-cause costs

- Mean PPPM all-cause costs were $16,478 and $2,801 in the on- and off-treatment phases, respectively, representing an 83% reduction in costs after treatment completion (**Figure 3**)
- Medication costs ($12,766) accounted for most of the total cost in the on-treatment phase, while medical costs ($1,959) were the primary drivers of the off-treatment costs (**Figure 3**)

Figure 3. Mean PPPM all-cause costs ($) between the on- versus off-treatment phases for patients in the R/R setting

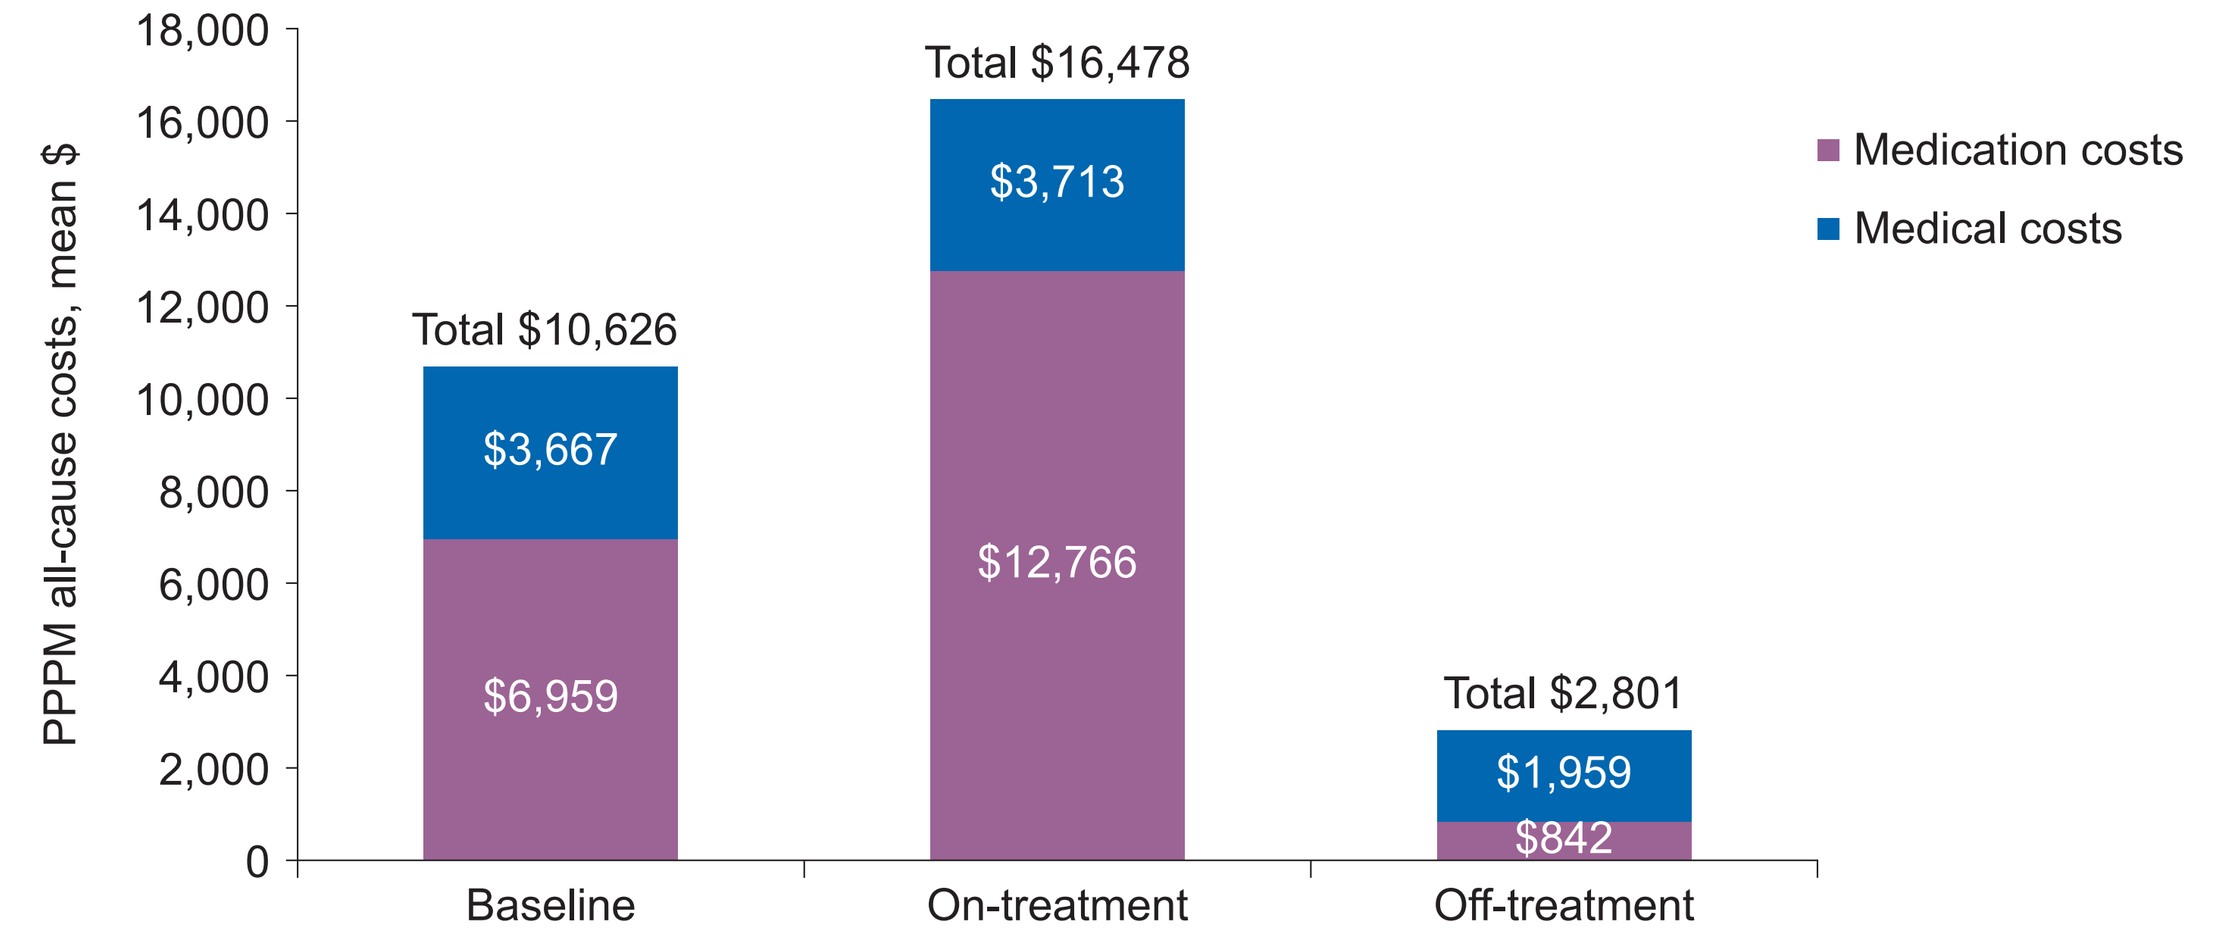

PPPM all-cause costs, mean $
18,000
16,000
14,000
12,000
10,000
8,000
6,000
4,000
2,000
0
Total $10,626
$3,667
$6,959
Total $16,478
$3,713
$12,766
Total $2,801
$1,959
$842
Baseline
On-treatment
Off-treatment
Medication costs
Medical costs

Individual components contributing to medical costs, mean (SD)

Medical costs	Baseline[a]	On-treatment	Off-treatment
Inpatient	$1,882 (3,636)	$1,461 (2,859)	$1,128 (4,762)
Outpatient	$1,398 (1,524)	$1,718 (1,622)	$548 (1,052)
Emergency room	$191 (366)	$226 (759)	$142 (487)

[a]Baseline entails the 12 months prior to VR initiation
PPPM, per patient per month; R/R, relapsed/refractory; SD, standard deviation

PPPM all-cause HRU

- Furthermore, the mean PPPM all-cause outpatient visits were higher in the on-treatment phase (3.2 vs 1.3)

Subsequent therapies

- Less than one quarter (20/108 [18.5%]) of VR-treated patients received subsequent treatment during the follow-up period (**Figure 4**)
- Of 35 patients who received 2L VR, only 8 received a subsequent line of therapy; of 44 patients who received 3L VR, only 5 received a subsequent line of therapy; of 24 patients who received VR as either 4L, 5L, or 6L, only 7 received a subsequent line of therapy (**Figure 4**)

Figure 4. Treatment sequencing for patients who received 2L–6L VR*

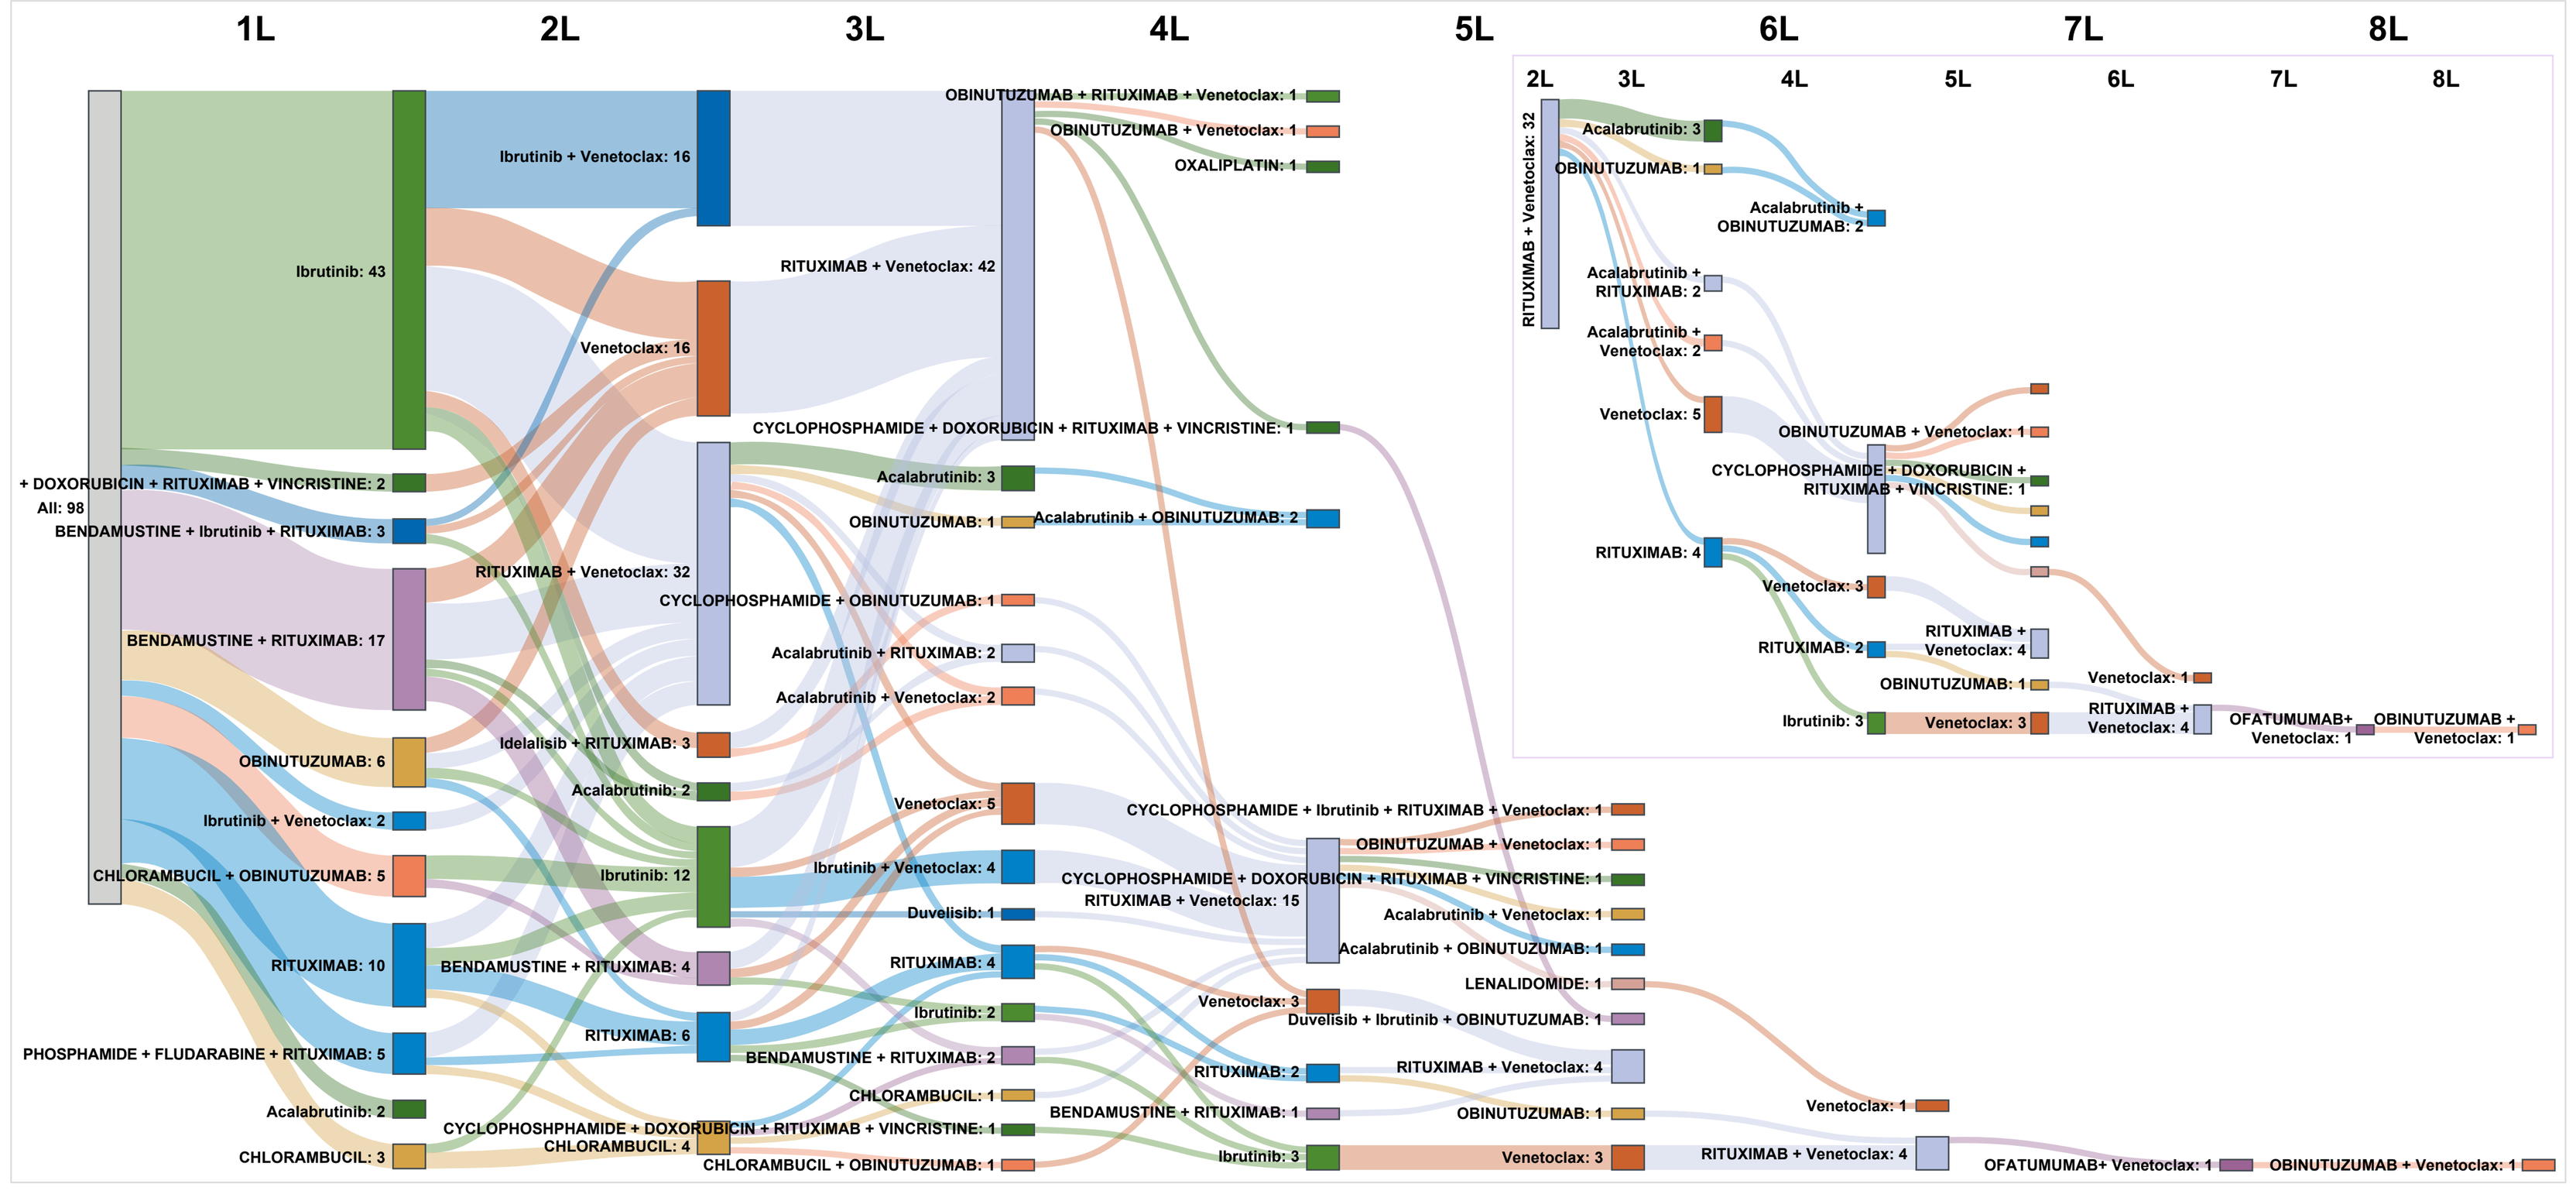

1L
2L
3L
4L
5L
6L
7L
8L
All: 98
Ibrutinib: 43
BENDAMUSTINE + Ibrutinib + RITUXIMAB: 3
BENDAMUSTINE + RITUXIMAB: 17
OBINUTUZUMAB: 6
Ibrutinib + Venetoclax: 2
CHLORAMBUCIL + OBINUTUZUMAB: 5
RITUXIMAB: 10
Acalabrutinib: 2
CHLORAMBUCIL: 3
Ibrutinib + Venetoclax: 16
Venetoclax: 16
RITUXIMAB + Venetoclax: 32
Idelalisib + RITUXIMAB: 3
Acalabrutinib: 2
Ibrutinib: 12
BENDAMUSTINE + RITUXIMAB: 4
RITUXIMAB: 6
CHLORAMBUCIL: 4
RITUXIMAB + Venetoclax: 42
OXALIPLATIN: 1
Acalabrutinib: 3
OBINUTUZUMAB: 1
Acalabrutinib + OBINUTUZUMAB: 2
CYCLOPHOSPHAMIDE + OBINUTUZUMAB: 1
Acalabrutinib + RITUXIMAB: 2
Acalabrutinib + Venetoclax: 2
Venetoclax: 5
Ibrutinib + Venetoclax: 4
Duvelisib: 1
RITUXIMAB: 4
Ibrutinib: 2
BENDAMUSTINE + RITUXIMAB: 2
CHLORAMBUCIL: 1
CHLORAMBUCIL + OBINUTUZUMAB: 1
RITUXIMAB + Venetoclax: 15
Venetoclax: 3
RITUXIMAB: 2
BENDAMUSTINE + RITUXIMAB: 1
Ibrutinib: 3
LENALIDOMIDE: 1
RITUXIMAB + Venetoclax: 4
OBINUTUZUMAB: 1
Venetoclax: 3
Venetoclax: 1
RITUXIMAB + Venetoclax: 4
OFATUMUMAB+ Venetoclax: 1
OBINUTUZUMAB + Venetoclax: 1

*ONLY 91% of the population are displayed – this was done to enable legibility of graph. 1L, first-line; 2L, second-line; 3L, third-line; 4L, fourth-line; 5L, fifth-line; 6L, sixth-line; 7L, seventh-line; 8L, eighth-line; VR, venetoclax plus rituximab

Study limitations

- As with any administrative claims-based study, coding errors are possible in the medical claims data
- The full economic benefits of off-treatment in this analysis were limited by the duration of the follow-up, and future analyses with longer off-treatment periods are warranted