NX-5948 and NX-2127 potently degrade a broad array of clinically-relevant BTK mutants that display resistance to inhibitors and other BTK degraders

Mark Noviski, Nivetha Brathaban, Stephanie Yung, Ratul Mukerji, Jordan Ye, Hugo Bousquet, Mateo Sanchez Garcia de los Rios, Brandon Bravo, Jeff Mihalic, Hao Lu, Cristiana Guiducci, Gwenn Hansen

Nurix Therapeutics, San Francisco, CA, USA

Abstract

Small molecule kinase inhibitors have revolutionized the treatment of chronic lymphocytic leukemia (CLL) by suppressing signaling pathways essential for tumor cell survival. Bruton's Tyrosine Kinase (BTK) inhibitors are widely used in the clinic for treatment of patients with CLL and other B cell malignancies. Acquired resistance mutations in BTK, however, can reduce or eliminate BTK inhibitor efficacy and represent a growing clinical challenge. Several BTK mutations have been reported in CLL patients. Mutations at C481 dramatically reduce the activity of covalent BTK inhibitors, whereas other clinically-observed mutations such as V416L, T474I, and L528W reduce or eliminate the activity of next-generation non-covalent inhibitors. While some of these mutations, such as V416L and L528W, abolish BTK kinase activity, they retain intact BCR signaling and BTK-dependent growth, indicating that kinase dead BTK mutants can elicit scaffold mediated signaling essential for malignant B cell survival.

To assess the impact of resistance mutations on the activity of BTK inhibitors, we generated several DLBCL lines (TMD8) harboring either BTK-C481S, C481R, V416L, T474I, or L528W point mutations. The C481S and C481R mutations eliminated the anti-proliferative effects of covalent inhibitors ibrutinib, acalabrutinib, and zanubrutinib, whereas these covalent inhibitors had variable effects on the V416L, T474I, and L528W mutants. By contrast, the non-covalent inhibitors pirtobrutinib, vecabrutinib, and fenebruitnib maintained potency against C481S but displayed partially reduced potency against the C481R mutation and dramatically reduced activity against the V416L, T474I, and L528W mutations. This variability in resistance mutant sensitivity to various BTK inhibitors complicates treatment decisions when patients relapse on BTK inhibitors, creating a therapeutic need for agents that can target resistance mutations more broadly. Targeted protein degradation represents one potentially mutation-agnostic therapeutic option since this modality's event-driven pharmacology requires only transient target interaction to promote complete elimination of a protein. This contrasts with inhibitors, which require continuous target occupancy to suppress protein activity and pathway activation, making them highly susceptible to point mutations that reduce drug affinity.

Here we assessed the activity of several chemically diverse heterobifunctional BTK degrader molecules against clinically observed BTK inhibitor-resistant mutations. All degrader molecules tested were able to degrade C481S- and C481R-mutant BTK and suppress the proliferation of TMD8 cells harboring these mutations. By contrast, many degrader molecules displayed marked loss of activity against the T474I, V416L, or L528W resistance mutations. By contrast, Nurix clinical stage degraders NX-5948 and NX-2127 were able to broadly target these resistance mutations. To determine the mechanism underlying the observed broad anti-mutant activity of our BTK degraders, we used surface plasmon resonance and FRET-based probe displacement assays to evaluate the binding of NX-5948 and NX-2127 to WT and mutant (C481S, C481R, T474I, V416L and L528W) BTK proteins. NX-5948 binds potently to BTK WT, C481S and T474I with single-digit nanomolar affinities, but loses 10-fold or greater binding affinity against the C481R, V416L, and L528W mutants. Furthermore, NX-2127 binds to some mutants with an affinity that would render most non-covalent BTK inhibitors inactive at therapeutically-relevant concentrations. Despite reduced BTK binary binding affinity, NX-5948 and NX-2127 induce potent degradation of all mutant forms of BTK and effectively suppress expression of activation markers and proliferation in TMD8 cells harboring these mutations. We propose that the positive cooperativity induced by NX-5948 and NX-2127 between BTK and the E3 ligase cereblon contributes to their potent and sustained degradation activity against BTK resistance mutants. The exceptional potency and activity of NX-5948 and NX-2127 against emerging BTK point mutations warrant their investigation in indications like CLL, that develop diverse resistance to inhibitor molecules. These degrader molecules may also have utility in earlier lines of therapy due to their ability to suppress scaffold-mediated BTK signaling. Phase 1a/b trials of NX-5948 and NX-2127 in patients with relapsed or refractory B-cell malignancies are ongoing (NX-5948: NCT05131022; NX-2127: NCT04830137).

Model

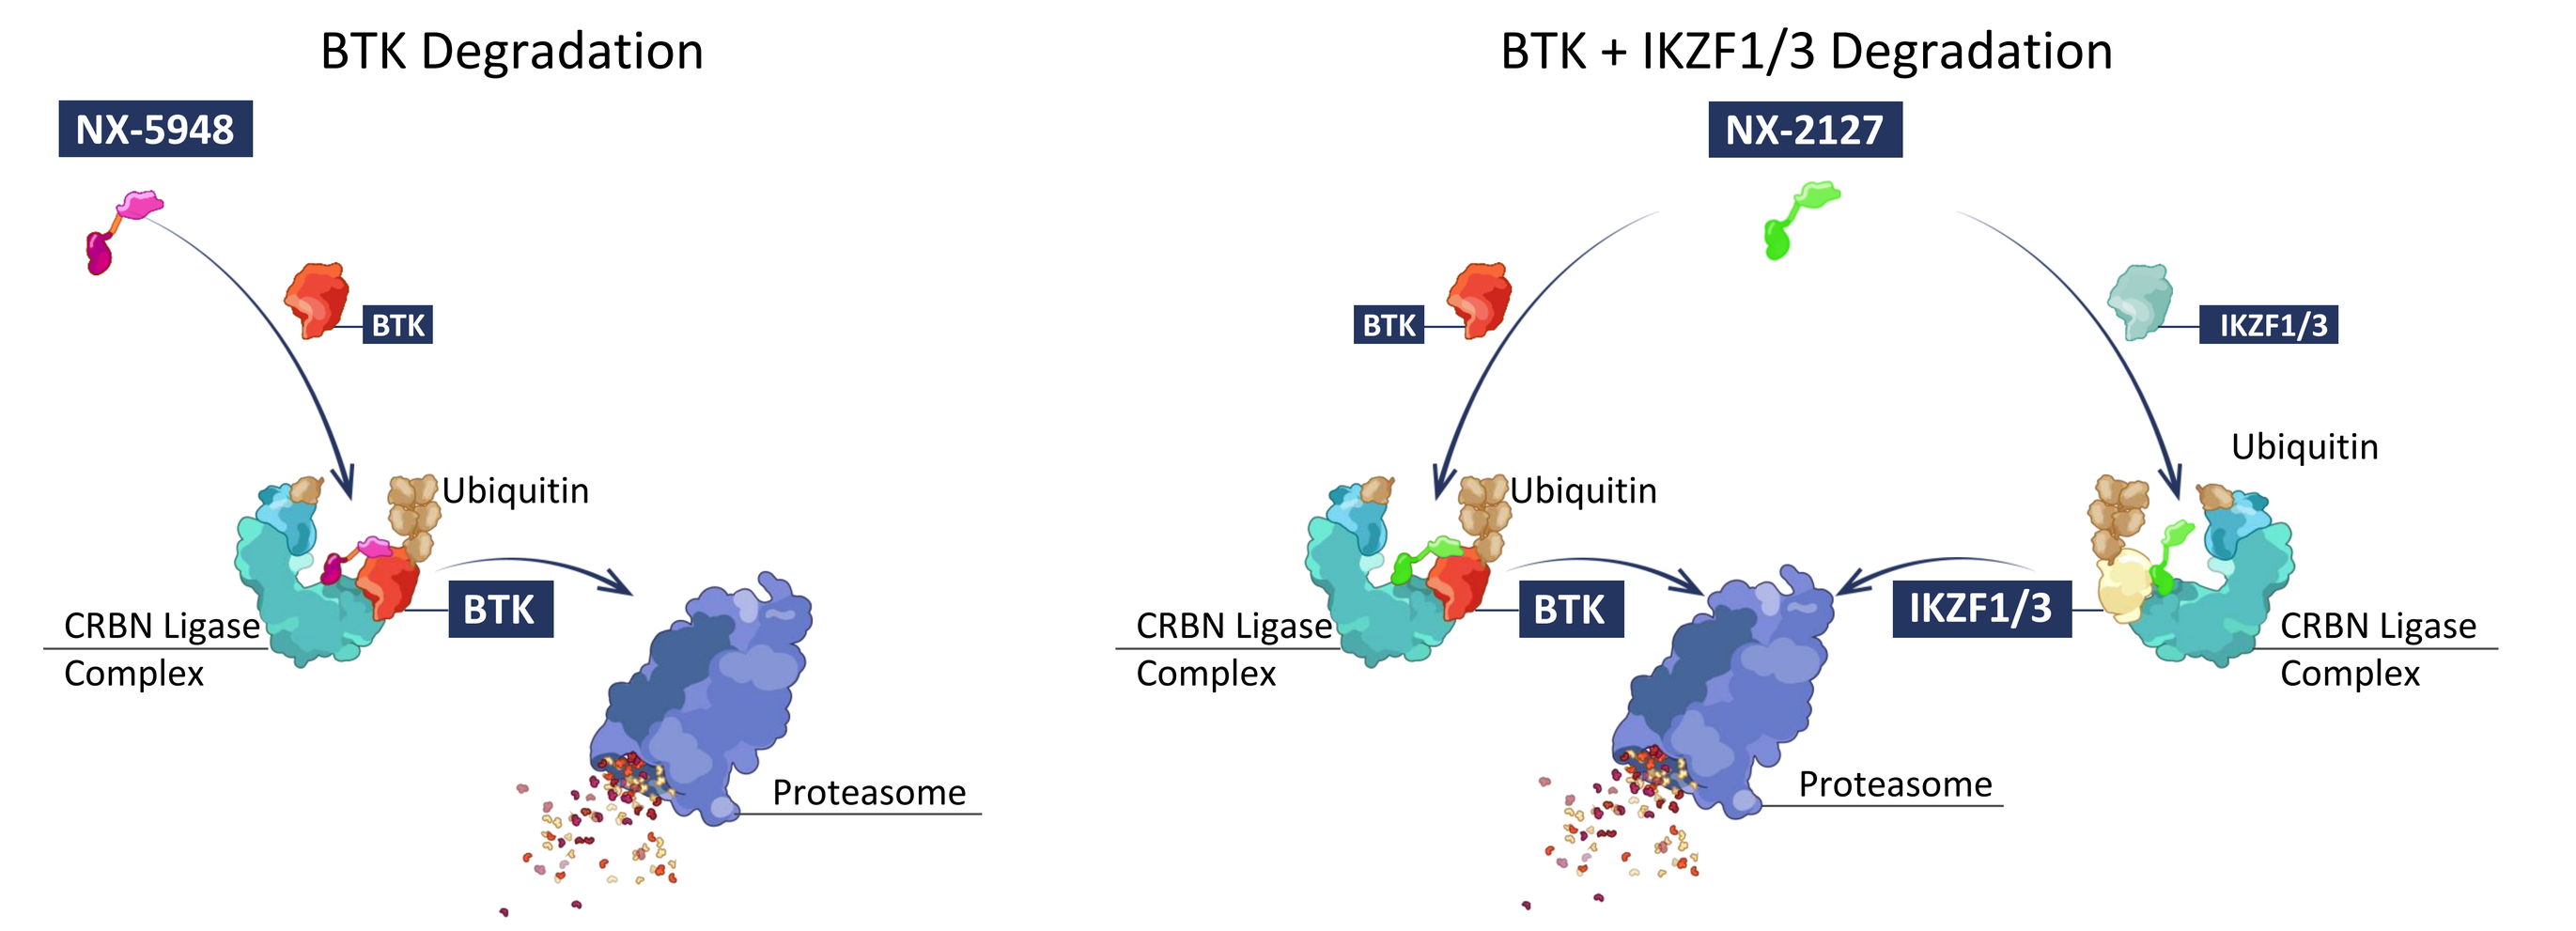
BTK Degradation
NX-5948
BTK
Ubiquitin
CRBN Ligase Complex
BTK
Proteasome
BTK + IKZF1/3 Degradation
NX-2127
BTK
IKZF1/3
Ubiquitin
Ubiquitin
CRBN Ligase Complex
BTK
IKZF1/3
CRBN Ligase Complex
Proteasome

Results

Figure 1: Acquired resistance mutations reduce or eliminate the anti-proliferative effects of BTK inhibitors

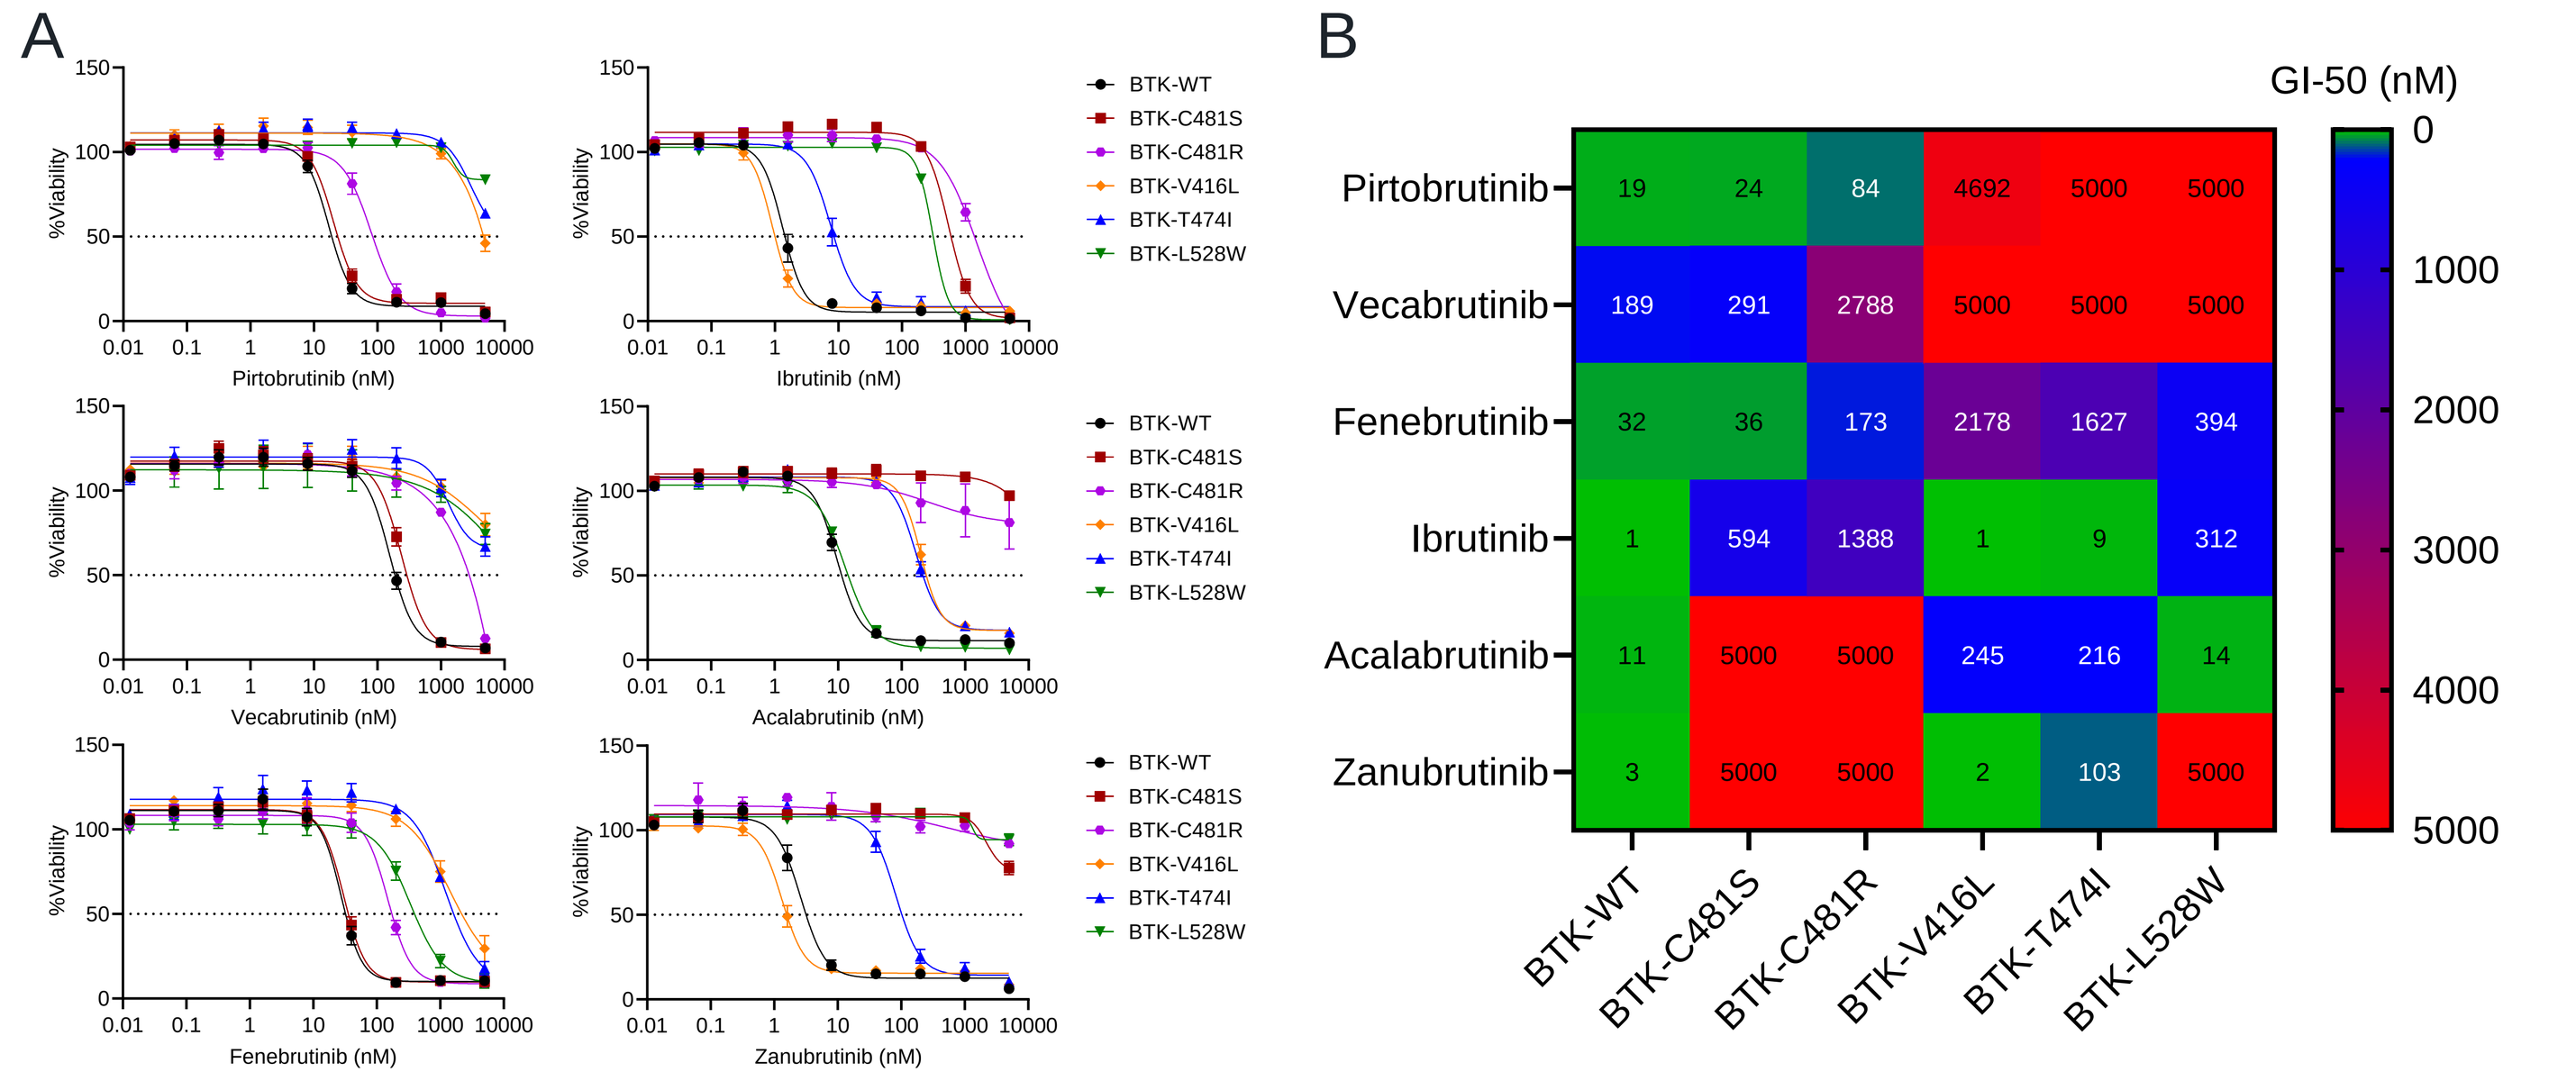
A
B
GI-50 (nM)
Pirtobrutinib 19 24 84 4692 5000 5000
Vecabrutinib 189 291 2788 5000 5000 5000
Fenebrutinib 32 36 173 2178 1627 394
Ibrutinib 1 594 1388 1 9 312
Acalabrutinib 11 5000 5000 245 216 14
Zanubrutinib 3 5000 5000 2 103 5000
BTK-WT BTK-C481S BTK-C481R BTK-V416L BTK-T474I BTK-L528W

(A) TMD8 cells harboring WT BTK or knock-in BTK mutations (C481S, C481R, V416L, T474I, or L528W) were incubated with non-covalent (left) and covalent (right) BTK inhibitors for 72 hours, and viability was assessed using CellTiter-Glo 2.0 (Promega). The percentage of viable cells in compound-treated samples was normalized to DMSO-treated controls. (C) GI_{50} values were calculated for compounds and lines in (A) and (B) and represent the concentration of compound that reduces cell growth by 50%. GI_{50} values were capped at the highest assay concentration (5000 nM) for compounds that did not achieve 50% killing. Curves and GI_{50} values are averaged from n $\geq$ 3 independent experiments. Mean $\pm$ SEM is displayed in (A) and (B).

Figure 2: NX-5948 and NX-2127 degrade mutant BTK and suppress activation markers in inhibitor-resistant TMD8 cells

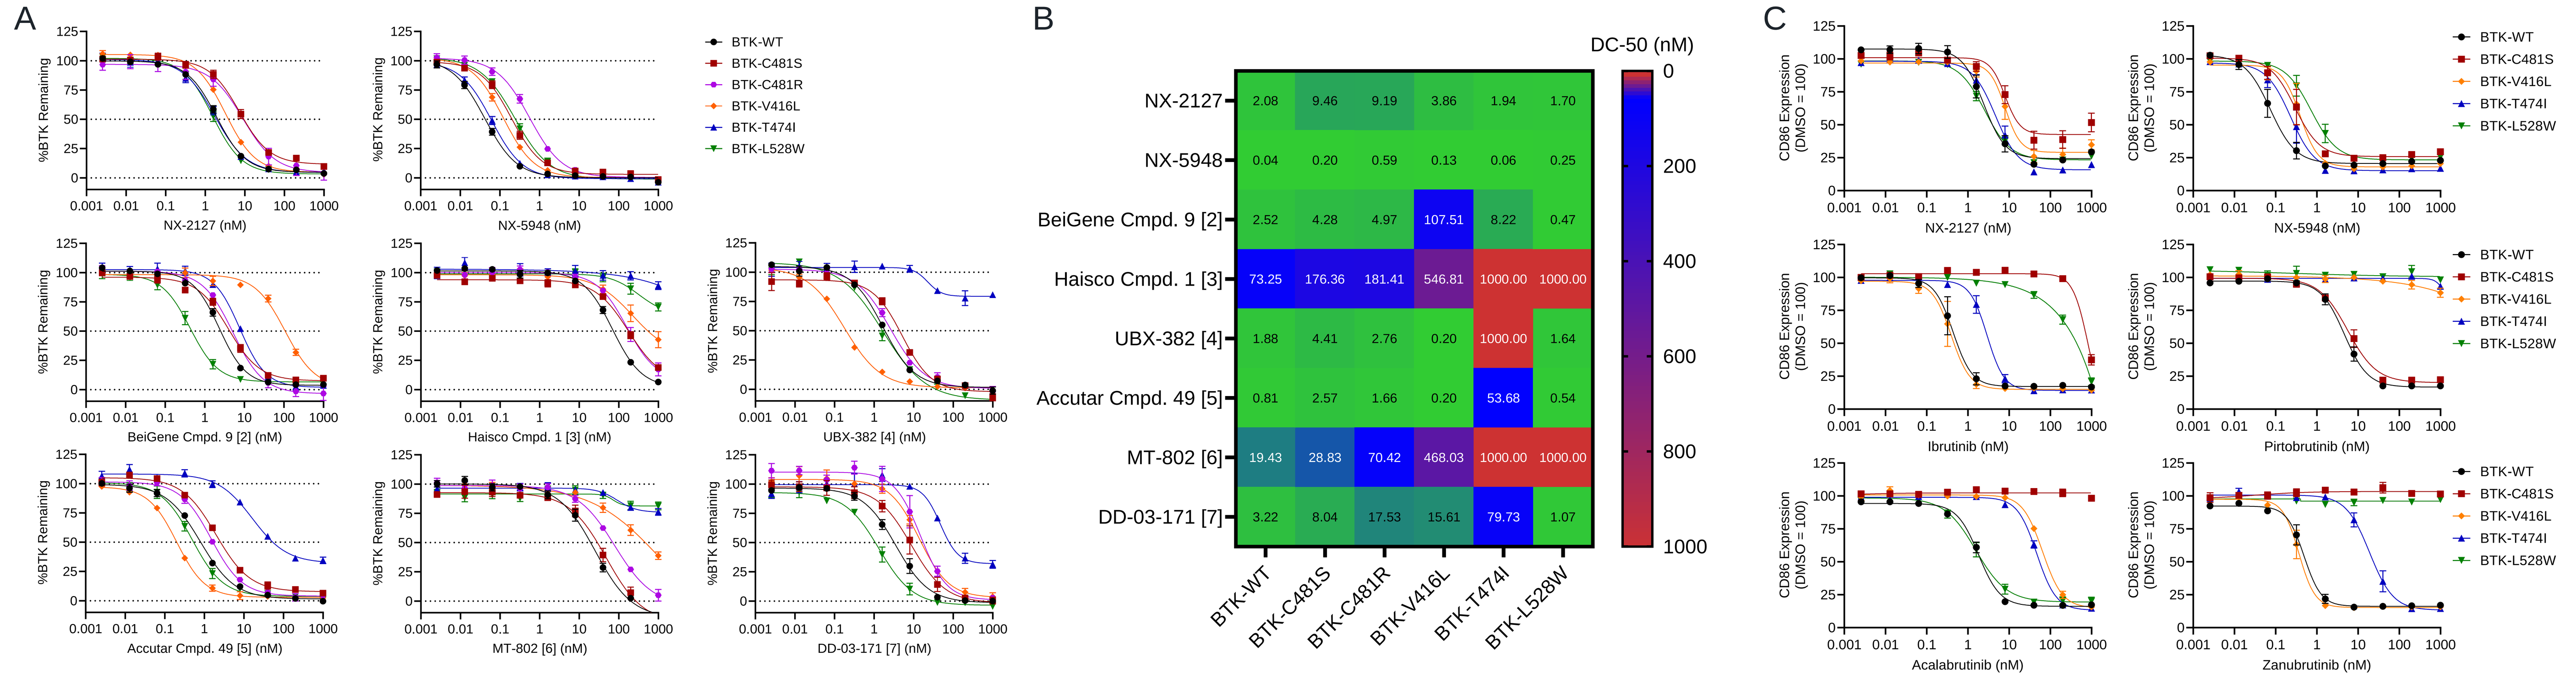
A
B
C
DC-50 (nM)
NX-2127 2.08 9.46 9.19 3.86 1.94 1.70
NX-5948 0.04 0.20 0.59 0.13 0.06 0.25
BeiGene Cmpd. 9 [2] 2.52 4.28 4.97 107.51 8.22 0.47
Haisco Cmpd. 1 [3] 73.25 176.36 181.41 546.81 1000.00 1000.00
UBX-382 [4] 1.88 4.41 2.76 0.20 1000.00 1.64
Accutar Cmpd. 49 [5] 0.81 2.57 1.66 0.20 53.68 0.54
MT-802 [6] 19.43 28.83 70.42 468.03 1000.00 1000.00
DD-03-171 [7] 3.22 8.04 17.53 15.61 79.73 1.07
BTK-WT BTK-C481S BTK-C481R BTK-V416L BTK-T474I BTK-L528W

(A) TMD8 cells harboring WT BTK or knock-in BTK mutations (C481S, C481R, V416L, T474I, or L528W) were incubated with BTK degraders for 24 hours, and BTK degradation was assessed by flow cytometry. (B) BTK DC_{50} values were calculated for degradation curves in (A) and correspond to the concentration of compound that promotes 50% degradation of BTK. (C) TMD8 cells harboring WT or mutant BTK were treated with NX-5948, NX-2127, or inhibitors for 24 hours, and surface CD86 expression was quantified by flow cytometry. Data were normalized to DMSO-treated controls and averaged from n $\geq$ 3 independent experiments. Mean $\pm$ SEM is displayed. Biomarker results were previously presented [8,9].

Figure 3. NX-5948 and NX-2127 demonstrate broad coverage of BTK inhibitor resistance mutations, in contrast to published BTK degraders

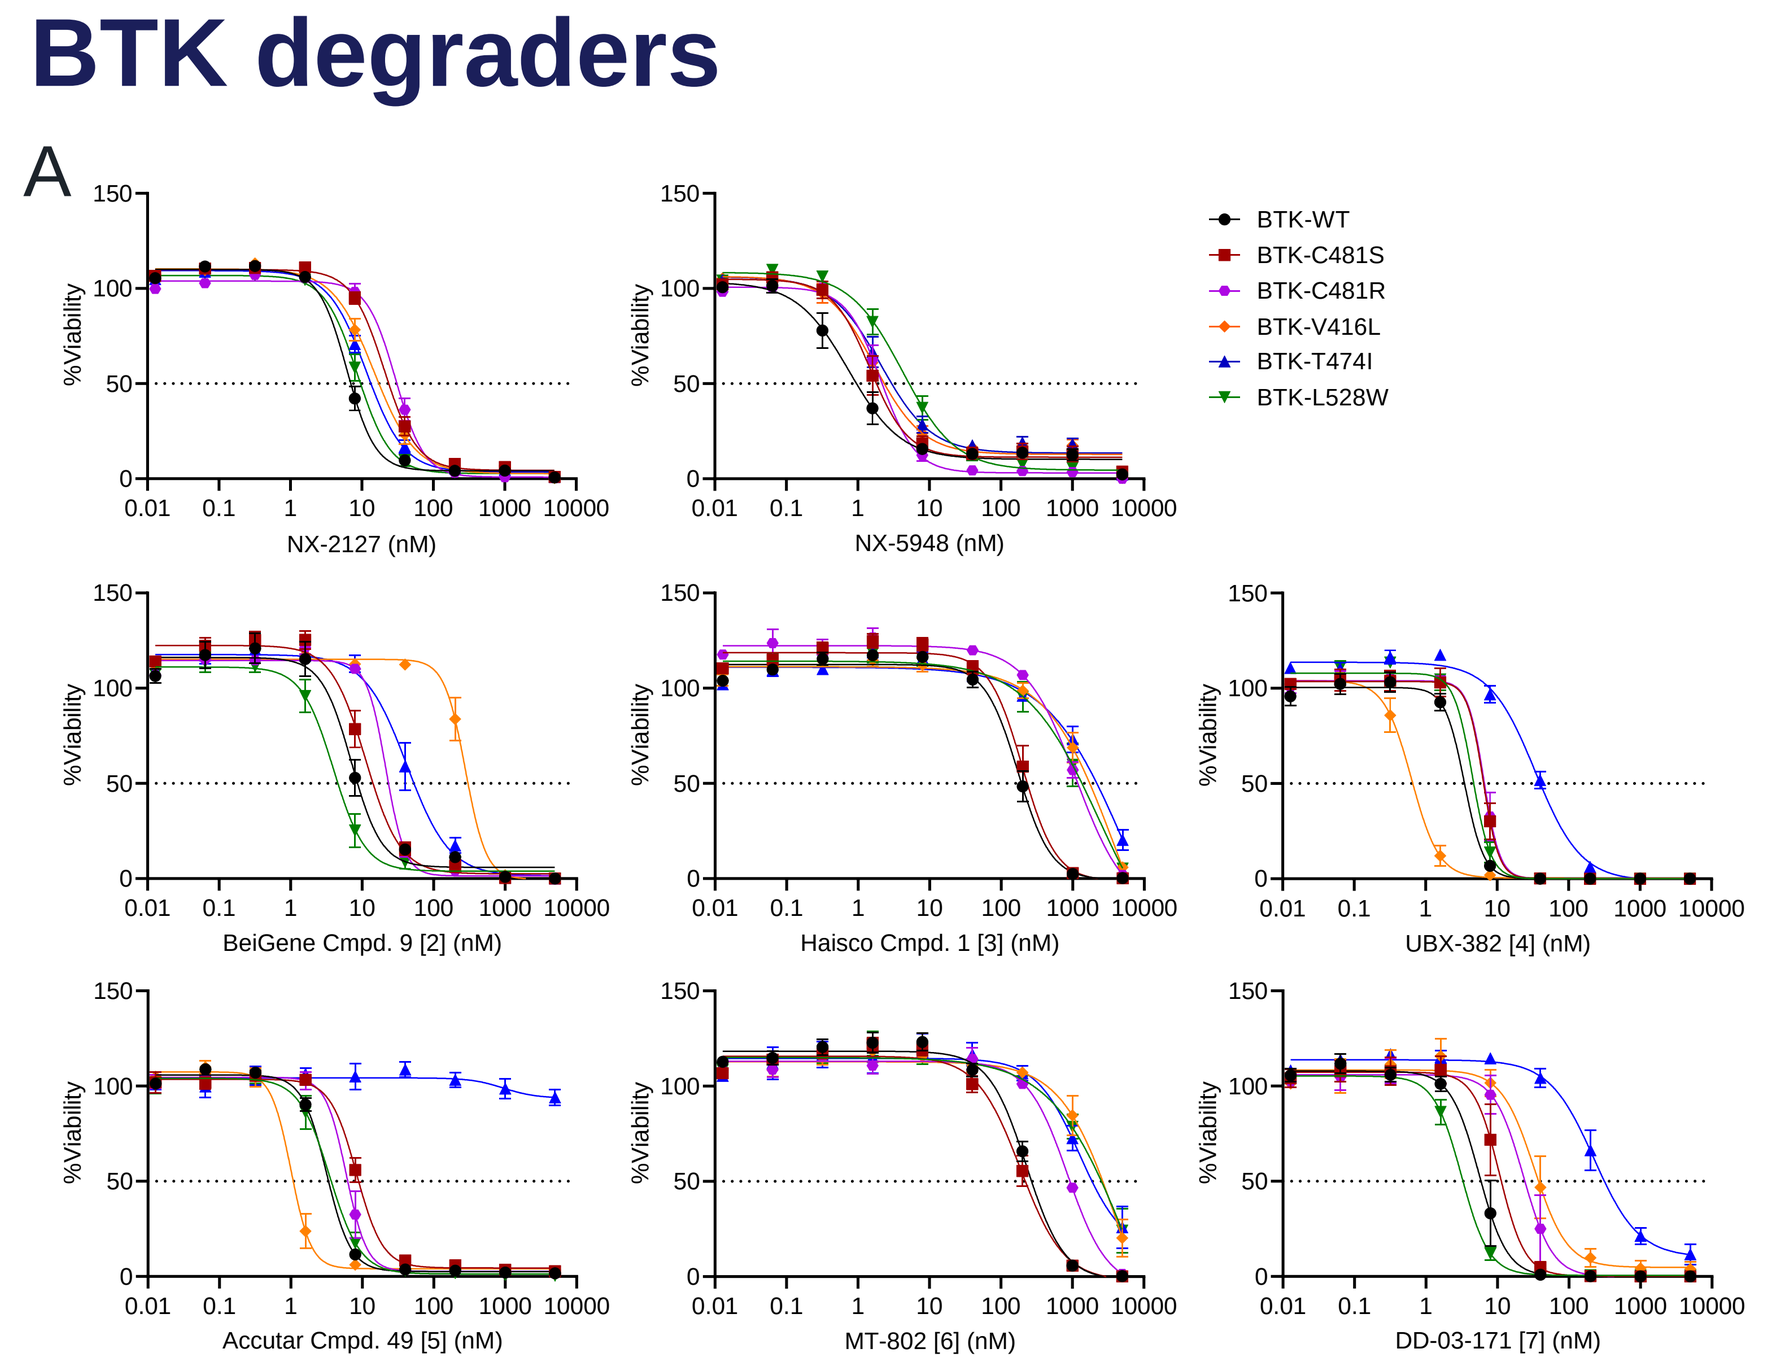
A
B
GI-50 (nM)
NX-2127 7 24 31 17 13 10
NX-5948 1 2 2 2 3 5
BeiGene Cmpd. 9 [2] 9 14 23 304 53 5
Haisco Cmpd. 1 [3] 193 232 1182 1734 2178 1349
UBX-382 [4] 4 6 7 1 40 5
Accutar Cmpd. 49 [5] 3 9 6 1 5000 4
MT-802 [6] 269 220 928 2665 1890 2557
DD-03-171 [7] 6 11 24 38 314 3
BTK-WT BTK-C481S BTK-C481R BTK-V416L BTK-T474I BTK-L528W

(A) TMD8 cells harboring WT BTK or knock-in BTK mutations were incubated with degrader molecules for 72 hours, and GI_{50} values (B) were calculated as in Figure 1. Mean $\pm$ SEM is displayed in (A), and data are averaged from n $\geq$ 3 independent experiments in (A) and (B).

Figure 4. Acquired resistance mutations reduce the binding of covalent and non-covalent BTK inhibitors to BTK

A

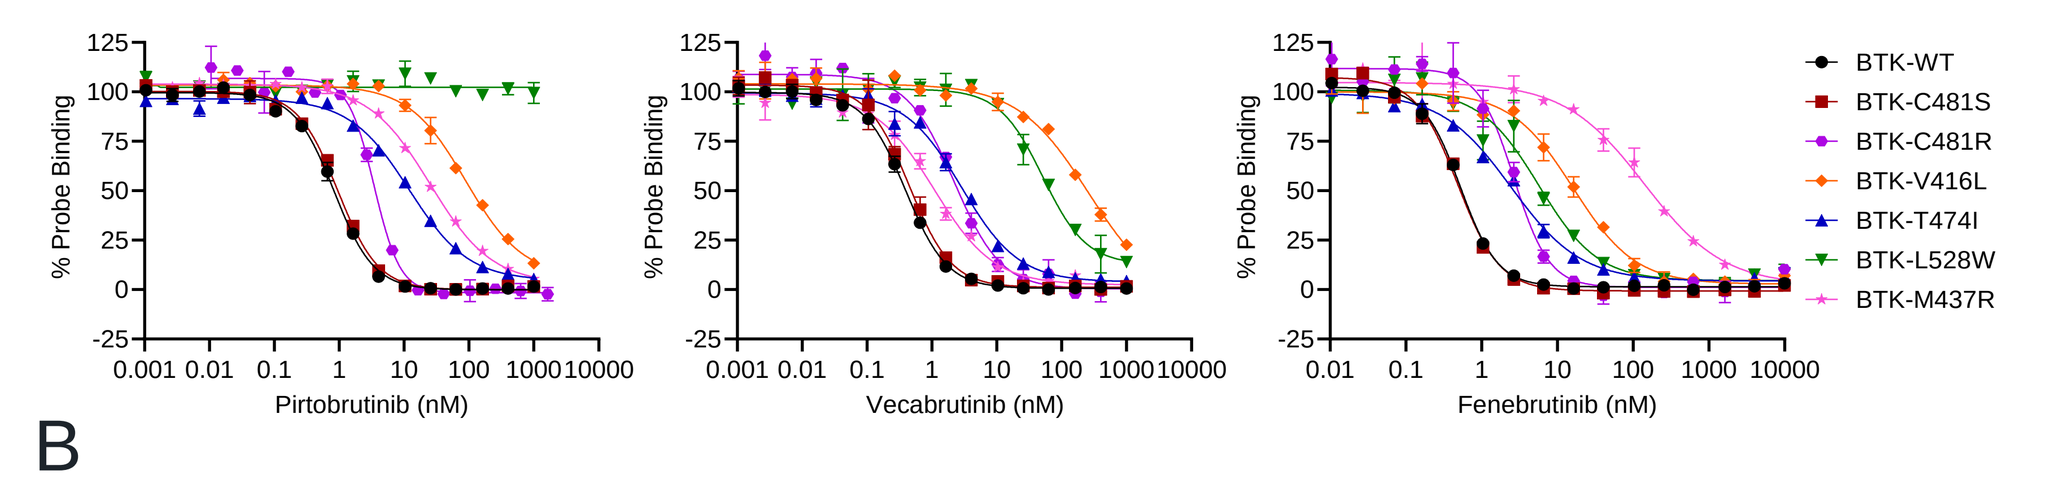

	$IC_{50} \pm$ SD (nM)		
	Pirtobrutinib	Vecabrutinib	Fenebrutinib
BTK-WT	0.76 ± 0.26	0.37 ± 0.05	0.70 ± 0.25
BTK-C481S	0.83 ± 0.30	0.43 ± 0.06	0.69 ± 0.28
BTK-C481R	2.69 ± 0.67	2.05 ± 0.21	3.19 ± 0.40
BTK-V416L	137 ± 71	147 ± 41	21 ± 4
BTK-T474I	13 ± 3	2.64 ± 0.34	2.74 ± 1.01
BTK-L528W	>1000	82 ± 19	7.60 ± 1.94
BTK-M437R	47 ± 24	1.00 ± 0.20	114 ± 36

B

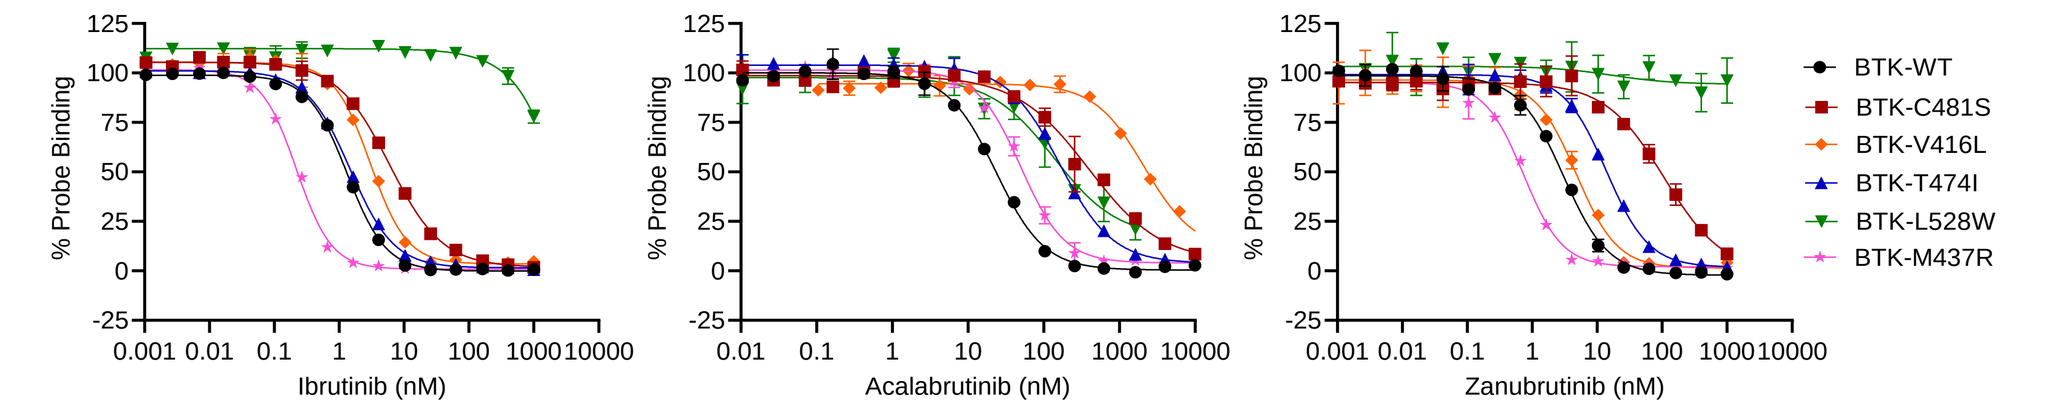

	$IC_{50} \pm$ SD (nM)		
	Ibrutinib	Acalabrutinib	Zanubrutinib
BTK-WT	1.4 ± 0.1	26.6 ± 2.4	2.7 ± 0.3
BTK-C481S	6.2 ± 0.3	467 ± 30	101 ± 6
BTK-V416L	3.8 ± 0.7	4101 ± 1331	4.7 ± 0.1
BTK-T474I	1.8 ± 0.3	189 ± 21	12.1 ± 1.5
BTK-L528W	>1000	240 ± 47	>1000
BTK-M437R	0.28 ± 0.07	52.7 ± 5.9	0.61 ± 0.11

(A-B) Representative biochemical probe-based TR-FRET competition assay data for non-covalent (A) and covalent (B) BTK inhibitors for WT and mutant BTK. Inhibitors were tested using 2.5-fold dilutions starting from 1 µM (or 10 µM for acalabrutinib and fenebrutinib). Average IC_{50} values for n $\geq$ 3 experiments are shown in tables. Probe displacement results (non-C481R) were previously presented [9].

Figure 5. NX-5948 and NX-2127 bind cooperatively with CRBN and BTK, overcoming observed reductions in binary binding affinity to some mutants

A

	NX-2127 binding to BTK by SPR (± SD)		
	K_D (nM)	k_{on} (10^6 $M^{-1}s^{-1}$)	k_{off} (s^{-1})
BTK-WT	18.3 ± 2.5	2.2 ± 1.6	0.039 ± 0.026
BTK-C481S	42.5 ± 7.3	1.9 ± 1.0	0.080 ± 0.037
BTK-V416L	102 ± 12	0.6 ± 0.3	0.065 ± 0.044
BTK-T474I	17.8 ± 2.2	2.3 ± 1.4	0.040 ± 0.024
BTK-L528W	89.1 ± 9.0	0.4 ± 0.1	0.034 ± 0.012
BTK-M437R	40.1 ± 10.2	1.0 ± 0.4	0.037 ± 0.015

	NX-5948 binding to BTK by SPR (± SD)		
	K_D (nM)	k_{on} (10^6 $M^{-1}s^{-1}$)	k_{off} (s^{-1})
BTK-WT	1.8 ± 0.4	5.9 ± 3.7	0.011 ± 0.009
BTK-C481S	5.9 ± 1.9	2.4 ± 0.2	0.014 ± 0.004
BTK-V416L	23 ± 1.7	3.5 ± 2.3	0.077 ± 0.046
BTK-T474I	2.1 ± 0.9	1.6 ± 1.1	0.004 ± 0.003
BTK-L528W	55.3 ± 7.5	2.8 ± 1.8	0.14 ± 0.09
BTK-M437R	3.5 ± 1.5	6.1 ± 2.6	0.019 ± 0.009

B

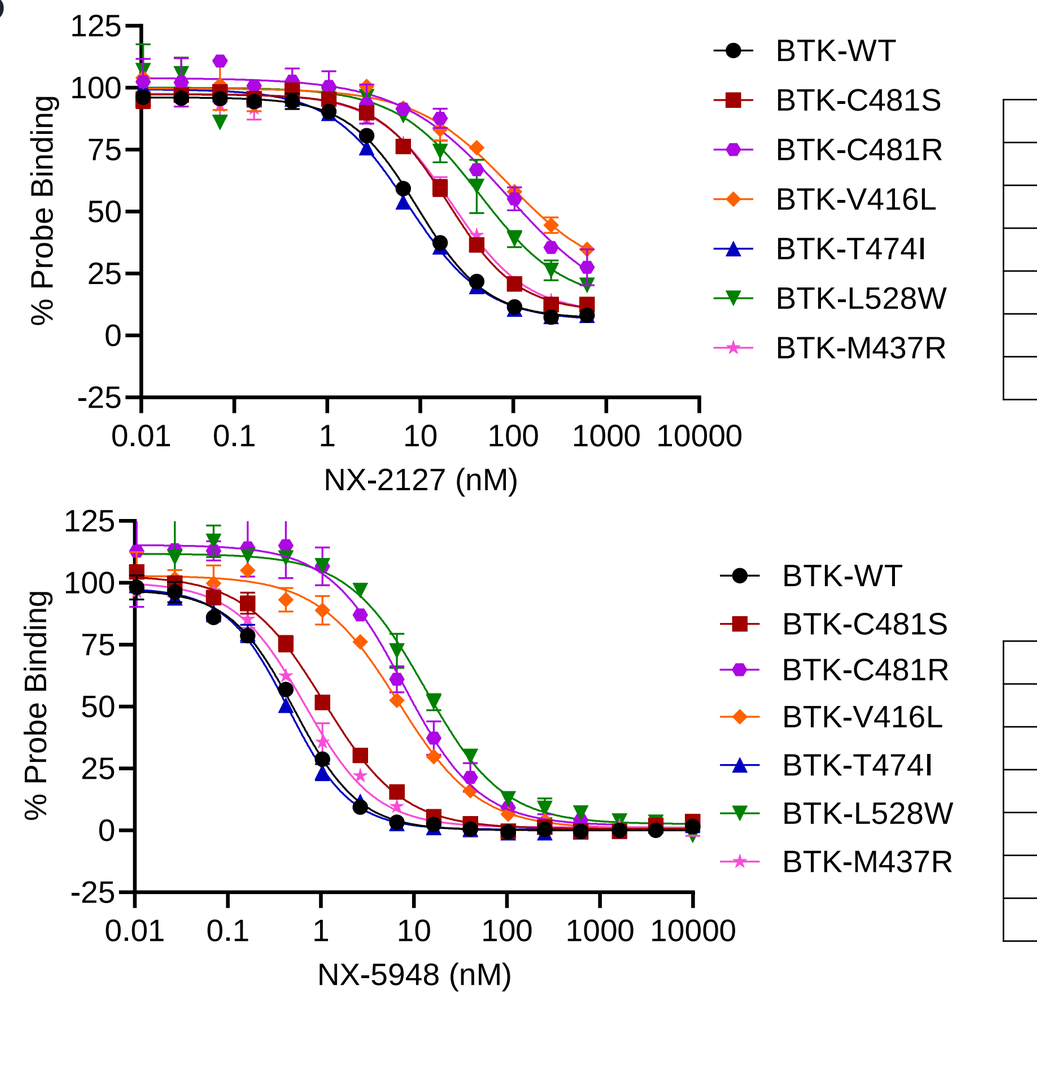

NX-2127	$IC_{50} \pm$ SD (nM) At Probe EC_{50}
BTK-WT	9 ± 1
BTK-C481S	19 ± 2
BTK-C481R	111 ± 29
BTK-V416L	169 ± 36
BTK-T474I	8 ± 2
BTK-L528W	72 ± 12
BTK-M437R	22 ± 2

NX-5948	$IC_{50} \pm$ SD (nM) At Probe EC_{50}
BTK-WT	0.74 ± 0.15
BTK-C481S	1.61 ± 0.38
BTK-C481R	7.88 ± 0.37
BTK-V416L	15 ± 6
BTK-T474I	0.62 ± 0.14
BTK-L528W	19 ± 5
BTK-M437R	1.06 ± 0.36

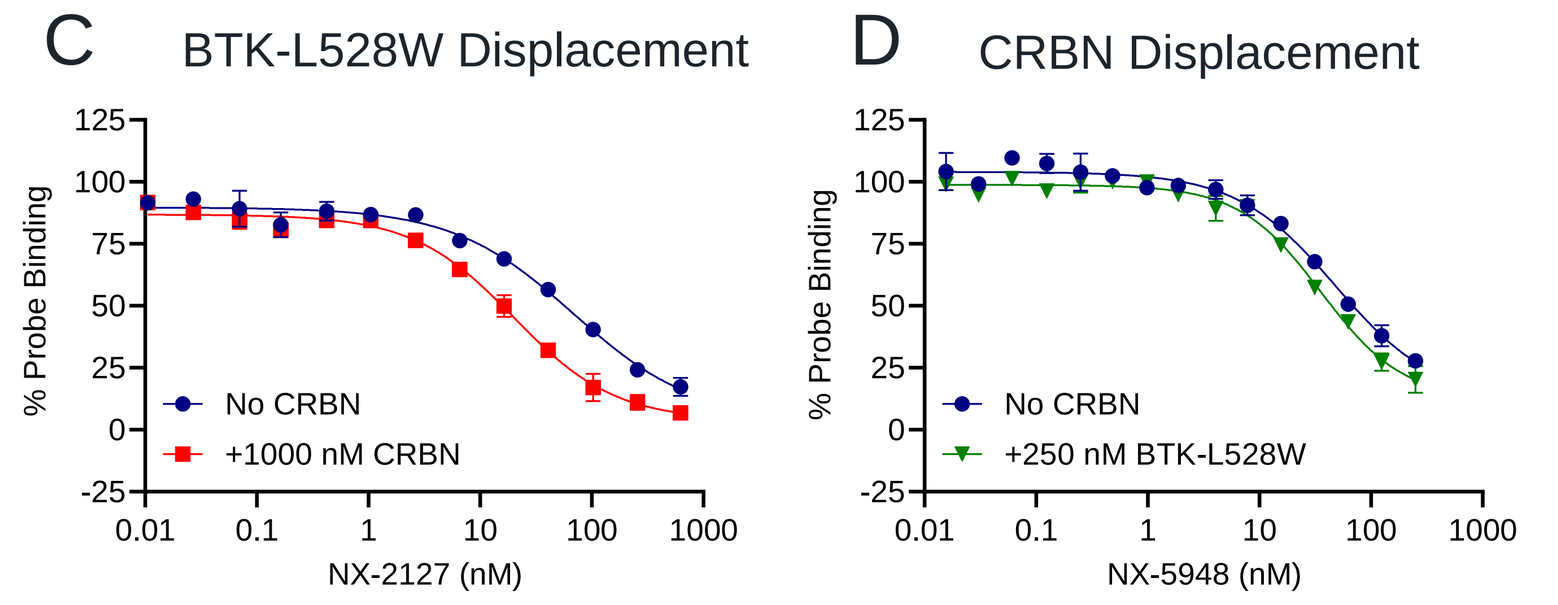
C BTK-L528W Displacement
No CRBN
+1000 nM CRBN
D CRBN Displacement
No CRBN
+250 nM BTK-L528W

E

	BTK Displacement at Probe EC_{50}			CRBN Displacement at Probe EC_{50}		
	NX-2127 IC_{50} (nM)			NX-5948 IC_{50} (nM)		
	No CRBN	1000 nM CRBN	Alpha	No BTK	250 nM BTK	Alpha
BTK-WT	10 ± 1	4.7 ± 1.5	2.2	46 ± 9	28 ± 2	1.6
BTK-C481S	22 ± 2	7.5 ± 1.8	3.0	48 ± 5	28 ± 2	1.7
BTK-V416L	165 ± 18	55 ± 8	3.0	57 ± 3	40 ± 4	1.4
BTK-T474I	8.6 ± 0.8	2.8 ± 0.7	3.1	44 ± 8	15.7 ± 0.6	2.8
BTK-L528W	70 ± 10	19 ± 5	3.7	48 ± 8	41.3 ± 0.9	1.2
BTK-M437R	23 ± 2	8.7 ± 3.2	2.7	58 ± 4	26.0 ± 0.28	2.2

(A) SPR binding parameters of NX-2127 and NX-5948 to WT and mutant BTK, reported as mean ± SD of n $\geq$ 3 independent measurements. Measurements were taken using a Biacore S200 instrument at 25°C, pH 7.5. (B) Representative biochemical probe-based TR-FRET competition assay data for NX-2127 and NX-5948 with BTK-WT and mutants. Compounds were tested using 2.5-fold dilutions starting from 10 or 1 µM at probe EC_{50}. SPR and probe displacement results (non-C481R) were previously presented [8,9]. (C) Representative biochemical probe-based TR-FRET cooperativity assay data for NX-2127 with BTK-L528W. NX-2127 was tested using 2.5-fold dilutions starting from 0.625 µM at probe EC50 in the absence and presence of saturating concentration of CRBN. (D) Additionally, NX-5948 was tested in a CRBN displacement assay using 2-fold dilutions starting at 0.25 µM at probe EC50 in the absence and presence of saturating concentration of BTK-L528W. (E) Summary statistics for experiments in (C) and (D). Average IC50 values from n = 2-3 independent experiments are shown in probe displacement tables. NX-2127 cooperativity results were previously presented [8].

Conclusions

- NX-5948 (DC-50 = 0.04 nM) and NX-2127 (DC-50 = 2.08 nM) promote potent degradation of BTK-WT
- Acquired resistance mutations reduce or abolish the anti-proliferative activity of BTK inhibitors. Small molecule inhibitors need to occupy the BTK active site continuously in order to suppress pathway activation and prevent tumor cell proliferation. Single amino acid changes in the active site are often sufficient to significantly reduce target occupancy.
- In contrast to occupancy-driven pharmacology, targeted protein degraders employ event-driven pharmacology, inducing ternary complexes with an E3 ligase to promote target ubiquitylation. Furthermore, the additional interactions between the target protein and E3 ligase can increase the stability of ternary complexes relative to binary complexes.
- NX-5948 and NX-2127 induce positive cooperativity between BTK and CRBN and retain the ability to degrade C481S, C481R, V416L, T474I, and L528W mutant BTK, irrespective of observed reduction in binary binding affinity to some mutants.
- NX-5948 and NX-2127 display superior coverage of novel BTKi resistance mutations compared to published BTK degraders, particularly in the context of T474I and V416L mutations.
- Downregulation of CD86 expression on TMD8 cells by inhibitors and degraders closely correlates with the anti-proliferative effects of NX-5948 and NX-2127.
- Phase 1a/b trials in patients with relapsed or refractory B-cell malignancies are ongoing for NX-5948 (NCT05131022) and NX-2127 (NCT04830137).

References

[1] Wang et al. 2022. N Engl J Med 386: 735-743.
[2] Example 9. WO 2021/219070 (BeiGene).
[3] Example 1. WO 2022/111449 (Haisco).
[4] Lim et al. 2023. Blood Adv. 7(1): 92-105.
[5] Example 49. WO 2022/169798 (Accutar)
[6] Buhimschi et al. 2018. Biochem. 57(26): 3564-75.
[7] Dobrovolsky et al. 2019. Blood 133(9): 952-961.
[8] Montoya et al. 2022. ASH Annual Meeting.
[9] Noviski et al. 2023. AACR Annual Meeting.

Disclosures

All authors are past or current employees of Nurix Therapeutics and hold company stock or stock options.

Presented at the International Workshop on CLL (iwCLL). October 6-9, 2023. Do not post.

Copies of this poster obtained through the Quick Response (QR) code are for personal use and may not be reproduced without permission from iwCLL and the author of this poster (mnoviski@nurixtx.com).

Poster Number: 2020