Poster 1552457

Longitudinal Review of Cardiac Events With Acalabrutinib in the Treatment of Chronic Lymphocytic Leukemia Using Data From Three Phase 3 Randomized Controlled Trials

Rupal O'Quinn,[1] Anthony J. Corry,[2] Naghmana Bajwa,[2] Suman Jannuru,[3] Hong Chen,[2] Katherine Stewart,[2] Jennifer R. Brown[4]

[1]Perelman School of Medicine, University of Pennsylvania, Philadelphia, PA, USA; [2]AstraZeneca, Gaithersburg, MD, USA; [3]AstraZeneca, South San Francisco, CA, USA; [4]Dana-Farber Cancer Institute, Boston, MA, USA

Objective

- To comprehensively analyze cardiac outcomes with acalabrutinib vs active comparators, including ibrutinib, in patients with and without CV disorders at baseline to assess the impact of CLL therapies

Conclusions

- The incidence of overall cardiac TEAEs across three phase 3 RCTs was numerically low overall with acalabrutinib compared with comparators
- The cardiac safety profile of acalabrutinib is similar across groups with and without CV disorders at baseline and in line with the known favorable safety profile of acalabrutinib
- This analysis does not suggest an increased risk of cardiac TEAEs and outcomes in acalabrutinib-treated patients, regardless of the presence of baseline CV disorders

Plain language summary

Why was this study done?
BTK inhibitors are effective treatments that target Bruton tyrosine kinase, a protein that plays a central role in CLL. Ibrutinib, which is the first approved BTK inhibitor, has been shown to be associated with risk of heart-related toxicities, including abnormal heart rhythms, heart failure, and sudden death. Acalabrutinib has less off-target binding to non-BTK kinases than ibrutinib, which may reduce off-target toxicities. We conducted this analysis to assess the impact of CLL therapies, including acalabrutinib, ibrutinib, and other anticancer drugs, on heart-related toxicities.

How were the data collected?
Data came from three major phase 3 randomized controlled clinical trials in CLL (ELEVATE-RR, ELEVATE-TN, and ASCEND) evaluating acalabrutinib vs other CLL treatments, which allowed for treatment comparisons. The exposure-adjusted incidence, which is the frequency of an event taking into account how long patients were receiving the drug, was calculated for heart-related side effects.

What were the results?
The results from the clinical trial data suggest a lower rate of heart-related toxicities overall with acalabrutinib compared with ibrutinib and other anticancer agents. The risk of heart-related toxicities was not increased with acalabrutinib, regardless of the number of heart diseases patients already had before receiving the treatment.

Why do the results matter to patients and physicians?
The results may help to inform treatment decisions for patients with CLL, particularly if they also have heart disease or are at higher risk of heart disease.

Poster

Supplementary material

Please scan this quick response (QR) code with your smartphone camera or app to obtain a copy of these materials.

Copies of this poster obtained through the QR code are for personal use and may not be reproduced without permission from iwCLL and the author of this poster.

Poster presented at the International Workshop on Chronic Lymphocytic Leukemia (iwCLL) Annual Meeting; October 6–9, 2023; Boston, MA

Corresponding author email address: Rupal.O'quinn@pennmedicine.upenn.edu

Introduction

- The first-generation BTKi ibrutinib demonstrated notable efficacy in CLL[1,2]
- However, ibrutinib has been shown to be associated with significant cardiac toxicity, including cardiac arrhythmias (eg, atrial fibrillation), cardiac failure, and sudden death[2]
- Acalabrutinib is a selective next-generation BTKi approved for the treatment of CLL with a more favorable CV safety profile with fewer atrial fibrillation events than ibrutinib[3,4]
- The CLL population is at higher risk for cardiac events due to advanced age, polypharmacy, significant comorbidities, and pre-existing cardiac disorders; thus, a comprehensive assessment of cardiac toxicities with CLL therapies is needed[5]

Methods

- Data were analyzed from three randomized phase 3 trials: ELEVATE-RR,[4] ELEVATE-TN,[6] and ASCEND[7] (**Figure 1**)
- 19 SMQs yielding ~2400 PTs were used to identify patients based on PMH and TEAEs (**Supplemental Table 1**)
- A subset of 89 PTs was used to identify the number of baseline CV disorders for additional analysis (**Supplemental Table 2**)

Figure 1. Analysis design

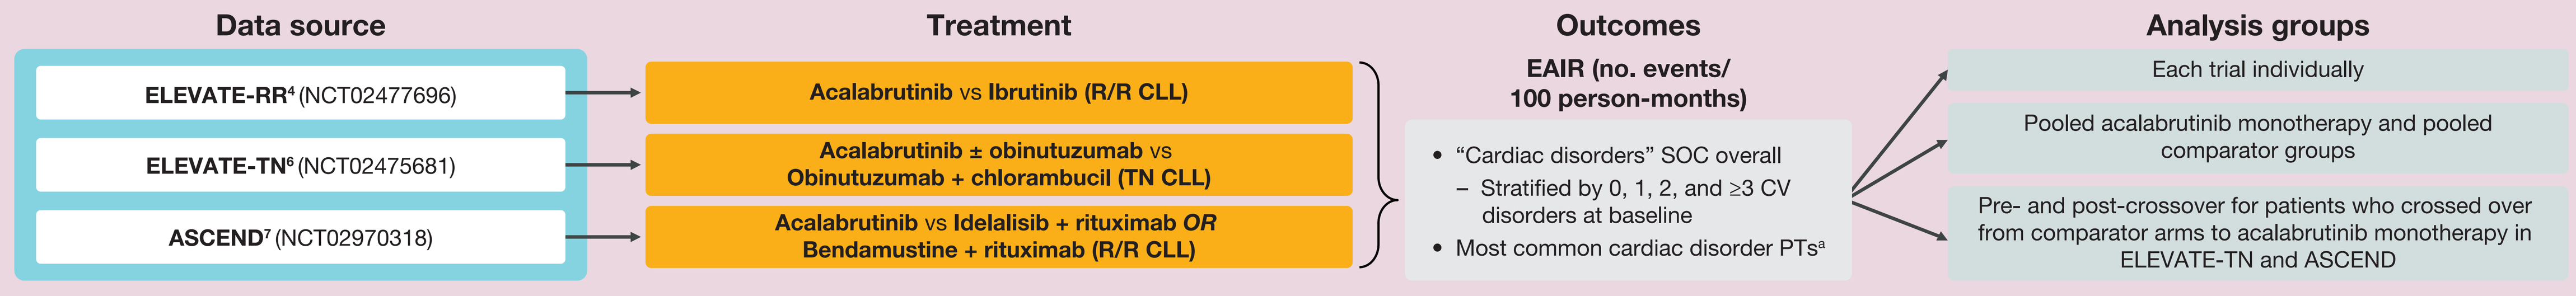

Data source
ELEVATE-RR[4] (NCT02477696)
ELEVATE-TN[6] (NCT02475681)
ASCEND[7] (NCT02970318)
Treatment
Acalabrutinib vs Ibrutinib (R/R CLL)
Acalabrutinib ± obinutuzumab vs Obinutuzumab + chlorambucil (TN CLL)
Acalabrutinib vs Idelalisib + rituximab OR Bendamustine + rituximab (R/R CLL)
Outcomes
EAIR (no. events/ 100 person-months)
"Cardiac disorders" SOC overall
– Stratified by 0, 1, 2, and ≥3 CV disorders at baseline
Most common cardiac disorder PTs[a]
Analysis groups
Each trial individually
Pooled acalabrutinib monotherapy and pooled comparator groups
Pre- and post-crossover for patients who crossed over from comparator arms to acalabrutinib monotherapy in ELEVATE-TN and ASCEND

[a]Analyzed only in the pooled acalabrutinib monotherapy and pooled comparator analysis group.

Results

Patients

- In total, 1362 patients with 3672 TEAEs were retrieved from the clinical trial database using the 19 SMQs; demographics and baseline characteristics were similar between acalabrutinib and comparator arms in all 3 trials (**Table 1**)
 - Of these patients, 404 (29.7%) had ≥1 baseline CV disorder (data by study provided in **Table 1**)
 - The distribution of baseline CV disorders was similar in the acalabrutinib and comparator arms across studies (**Table 1**)
 - 599 patients were treated with acalabrutinib monotherapy
 - 178 were treated with acalabrutinib + obinutuzumab
 - 585 were treated with comparators (including ibrutinib and other anticancer agents)
 - 72 received obinutuzumab + chlorambucil and crossed over to acalabrutinib monotherapy in ELEVATE-TN
 - 80 received idelalisib + rituximab OR bendamustine + rituximab and crossed over to acalabrutinib monotherapy in ASCEND
- Median exposure with BTKis (given continuously) was longer than with comparators across studies (**Table 1**)

Cardiac disorder events in individual trials

- In the 3 RCTs, the overall EAIR of cardiac disorder events of any grade was numerically lower in the acalabrutinib arm than in the comparator arms (**Figure 2A**)
 - No specific trend was seen among patients with ≥1 baseline CV disorder; however, the number of events was limited (**Figure 2B-D**)
- EAIR of any grade de novo cardiac disorder events (in patient with 0 baseline CV disorders) is shown for each study in **Figure 2B-D** and described below:
 - ELEVATE-RR: EAIR with ibrutinib was twice that seen with acalabrutinib (0.67 vs 0.34)
 - ELEVATE-TN: EAIR was numerically lower with acalabrutinib + obinutuzumab (0.28) and acalabrutinib monotherapy (0.25) than with chlorambucil + obinutuzumab (0.59)
 - ASCEND: EAIR was numerically lower with acalabrutinib (0.28) than with idelalisib + rituximab (0.44) and bendamustine + rituximab (0.54)

Cardiac disorder events in pooled acalabrutinib and pooled comparator groups

- EAIR of cardiac disorder events of any grade was approximately twice as high for pooled comparator as for pooled acalabrutinib (**Figure 3**)
 - EAIR of fatal events was 3–4 times higher for pooled comparator vs pooled acalabrutinib
- The most frequent cardiac disorder PT in both groups was atrial fibrillation (**Table 2**)
- EAIR of cardiac disorder events in patients with no baseline CV disorders was numerically lower for pooled acalabrutinib vs pooled comparator (**Table 3**)
 - EAIR of fatal events was 3 times higher for pooled comparator vs pooled acalabrutinib in patients with no baseline CV disorders
 - EAIR of fatal events did not increase in patients with ≥1 baseline CV disorder vs no baseline CV disorders, and was low overall

Cardiac disorder events in crossover patients (ELEVATE-TN and ASCEND)

- In ELEVATE-TN (n=72), EAIR of any-grade cardiac disorder events was lower during the post-crossover period with acalabrutinib than during the pre-crossover period with comparator treatment overall (**Figure 4**)
 - EAIR of grade ≥3 events was similar pre- and post-crossover, and only 1 fatal event was observed, which occurred in the post-crossover period
 - Among patients with no CV disorders at baseline, EAIR of cardiac disorder events was similar pre- and post-crossover; among those with ≥1 CV disorder at baseline, EAIR of cardiac disorder events was higher pre- vs post-crossover (**Table 4**)
- In ASCEND (n=80), EAIR of any-grade cardiac disorder events was low before and after crossover to acalabrutinib (**Figure 4**)
 - EAIR of grade ≥3 events was numerically higher post-crossover vs pre-crossover; no fatal events were observed
 - Results were similar pre- and post-crossover regardless of the number of CV disorders at baseline (**Table 4**)

Table 1. Patient demographics, baseline characteristics, and treatment exposure

Characteristic	ELEVATE-RR Acala (n=266)	ELEVATE-RR Ibr (n=263)	ELEVATE-TN Acala + Obin (n=178)	ELEVATE-TN Acala (n=179)	ELEVATE-TN Obin + Clb (n=169)	ASCEND Acala (n=154)	ASCEND Idela + Ritux (n=118)	ASCEND Benda + Ritux (n=35)
Age, median (range), years	66 (41–89)	65 (28–88)	70 (41–88)	70 (44–87)	71 (46–91)	68 (32–89)	67 (34–88)	68 (53–90)
Age group, n (%)								
≥65 years	142 (53.4)	142 (54.0)	143 (80.3)	151 (84.4)	147 (87.0)	97 (63.0)	73 (61.9)	25 (71.4)
≥75 years	44 (16.5)	42 (16.0)	53 (29.8)	50 (27.9)	50 (30.0)	34 (22.1)	23 (19.5)	8 (22.9)
Male, n (%)	184 (69.0)	192 (73.0)	111 (62.4)	110 (61.5)	103 (61.0)	107 (69.5)	77 (65.3)	22 (62.9)
BMI ≥30, n (%)	54 (20.3)	64 (24.3)	48 (27.0)	49 (27.4)	42 (24.9)	38 (24.7)	27 (22.9)	8 (22.9)
Current or former smoker >10 years, n (%)	93 (35.0)	87 (33.1)	56 (31.5)	52 (29.1)	66 (39.1)	N/A	N/A	N/A
Number of CV disorders at baseline, n (%)								
0	190 (71.4)	195 (74.1)	119 (66.9)	125 (69.8)	113 (66.9)	111 (72.1)	84 (71.2)	21 (60.0)
1	47 (17.7)	43 (16.3)	40 (22.5)	39 (21.8)	35 (20.7)	25 (16.2)	20 (16.9)	9 (25.7)
2	15 (5.6)	15 (5.7)	12 (6.7)	5 (2.8)	12 (7.1)	8 (5.2)	9 (7.6)	3 (8.6)
≥3	14 (5.3)	10 (3.8)	7 (3.9)	10 (5.6)	9 (5.3)	10 (6.5)	5 (4.2)	2 (5.7)
Baseline history, n (%)								
Hypertension	129 (48.5)	128 (48.7)	106 (59.6)	112 (62.6)	115 (68.1)	84 (54.6)	55 (46.6)	24 (68.6)
Diabetes	53 (19.9)	49 (18.6)	39 (21.9)	31 (17.3)	36 (21.3)	34 (22.1)	18 (15.3)	5 (14.3)
Myocardial infarction	10 (3.8)	14 (5.3)	9 (5.1)	4 (2.2)	7 (4.1)	3 (2.0)	6 (5.1)	0
Chronic kidney disease	20 (7.5)	19 (7.2)	12 (6.7)	15 (8.4)	13 (7.7)	7 (4.6)	5 (4.2)	0
Arrhythmias	40 (15.0)	32 (12.2)	32 (18.0)	24 (13.4)	22 (13.0)	23 (14.9)	18 (15.3)	5 (14.3)
Cerebrovascular/ transient ischemic attack	12 (4.5)	12 (4.6)	6 (3.4)	10 (5.6)	9 (5.3)	9 (5.8)	7 (5.9)	1 (2.9)
Coronary artery disease	27 (10.2)	29 (11.0)	18 (10.1)	21 (11.7)	21 (12.4)	23 (14.9)	15 (12.7)	9 (25.7)
Hyperlipidemia (PT)	22 (8.3)	11 (4.2)	16 (9.0)	22 (12.3)	25 (14.8)	8 (5.2)	5 (4.2)	5 (14.3)
Heart failure	7 (2.6)	9 (3.4)	7 (3.9)	4 (2.2)	5 (3.0)	6 (3.9)	5 (4.2)	2 (5.7)
Treatment duration, months	Median 38.3	Median 35.5	Median 58	Median 58	Maximum 6[a]	Median 44.2	Idela: median 11.5 Ritux: median 5.5	Benda: maximum 6[a] Ritux: median 5.5

[a]6 cycles (28-day cycles).

Figure 2. EAIR of treatment-emergent cardiac disorder events (SOC) in phase 3 trials overall (A) and by number of CV disorders at baseline (B-D)

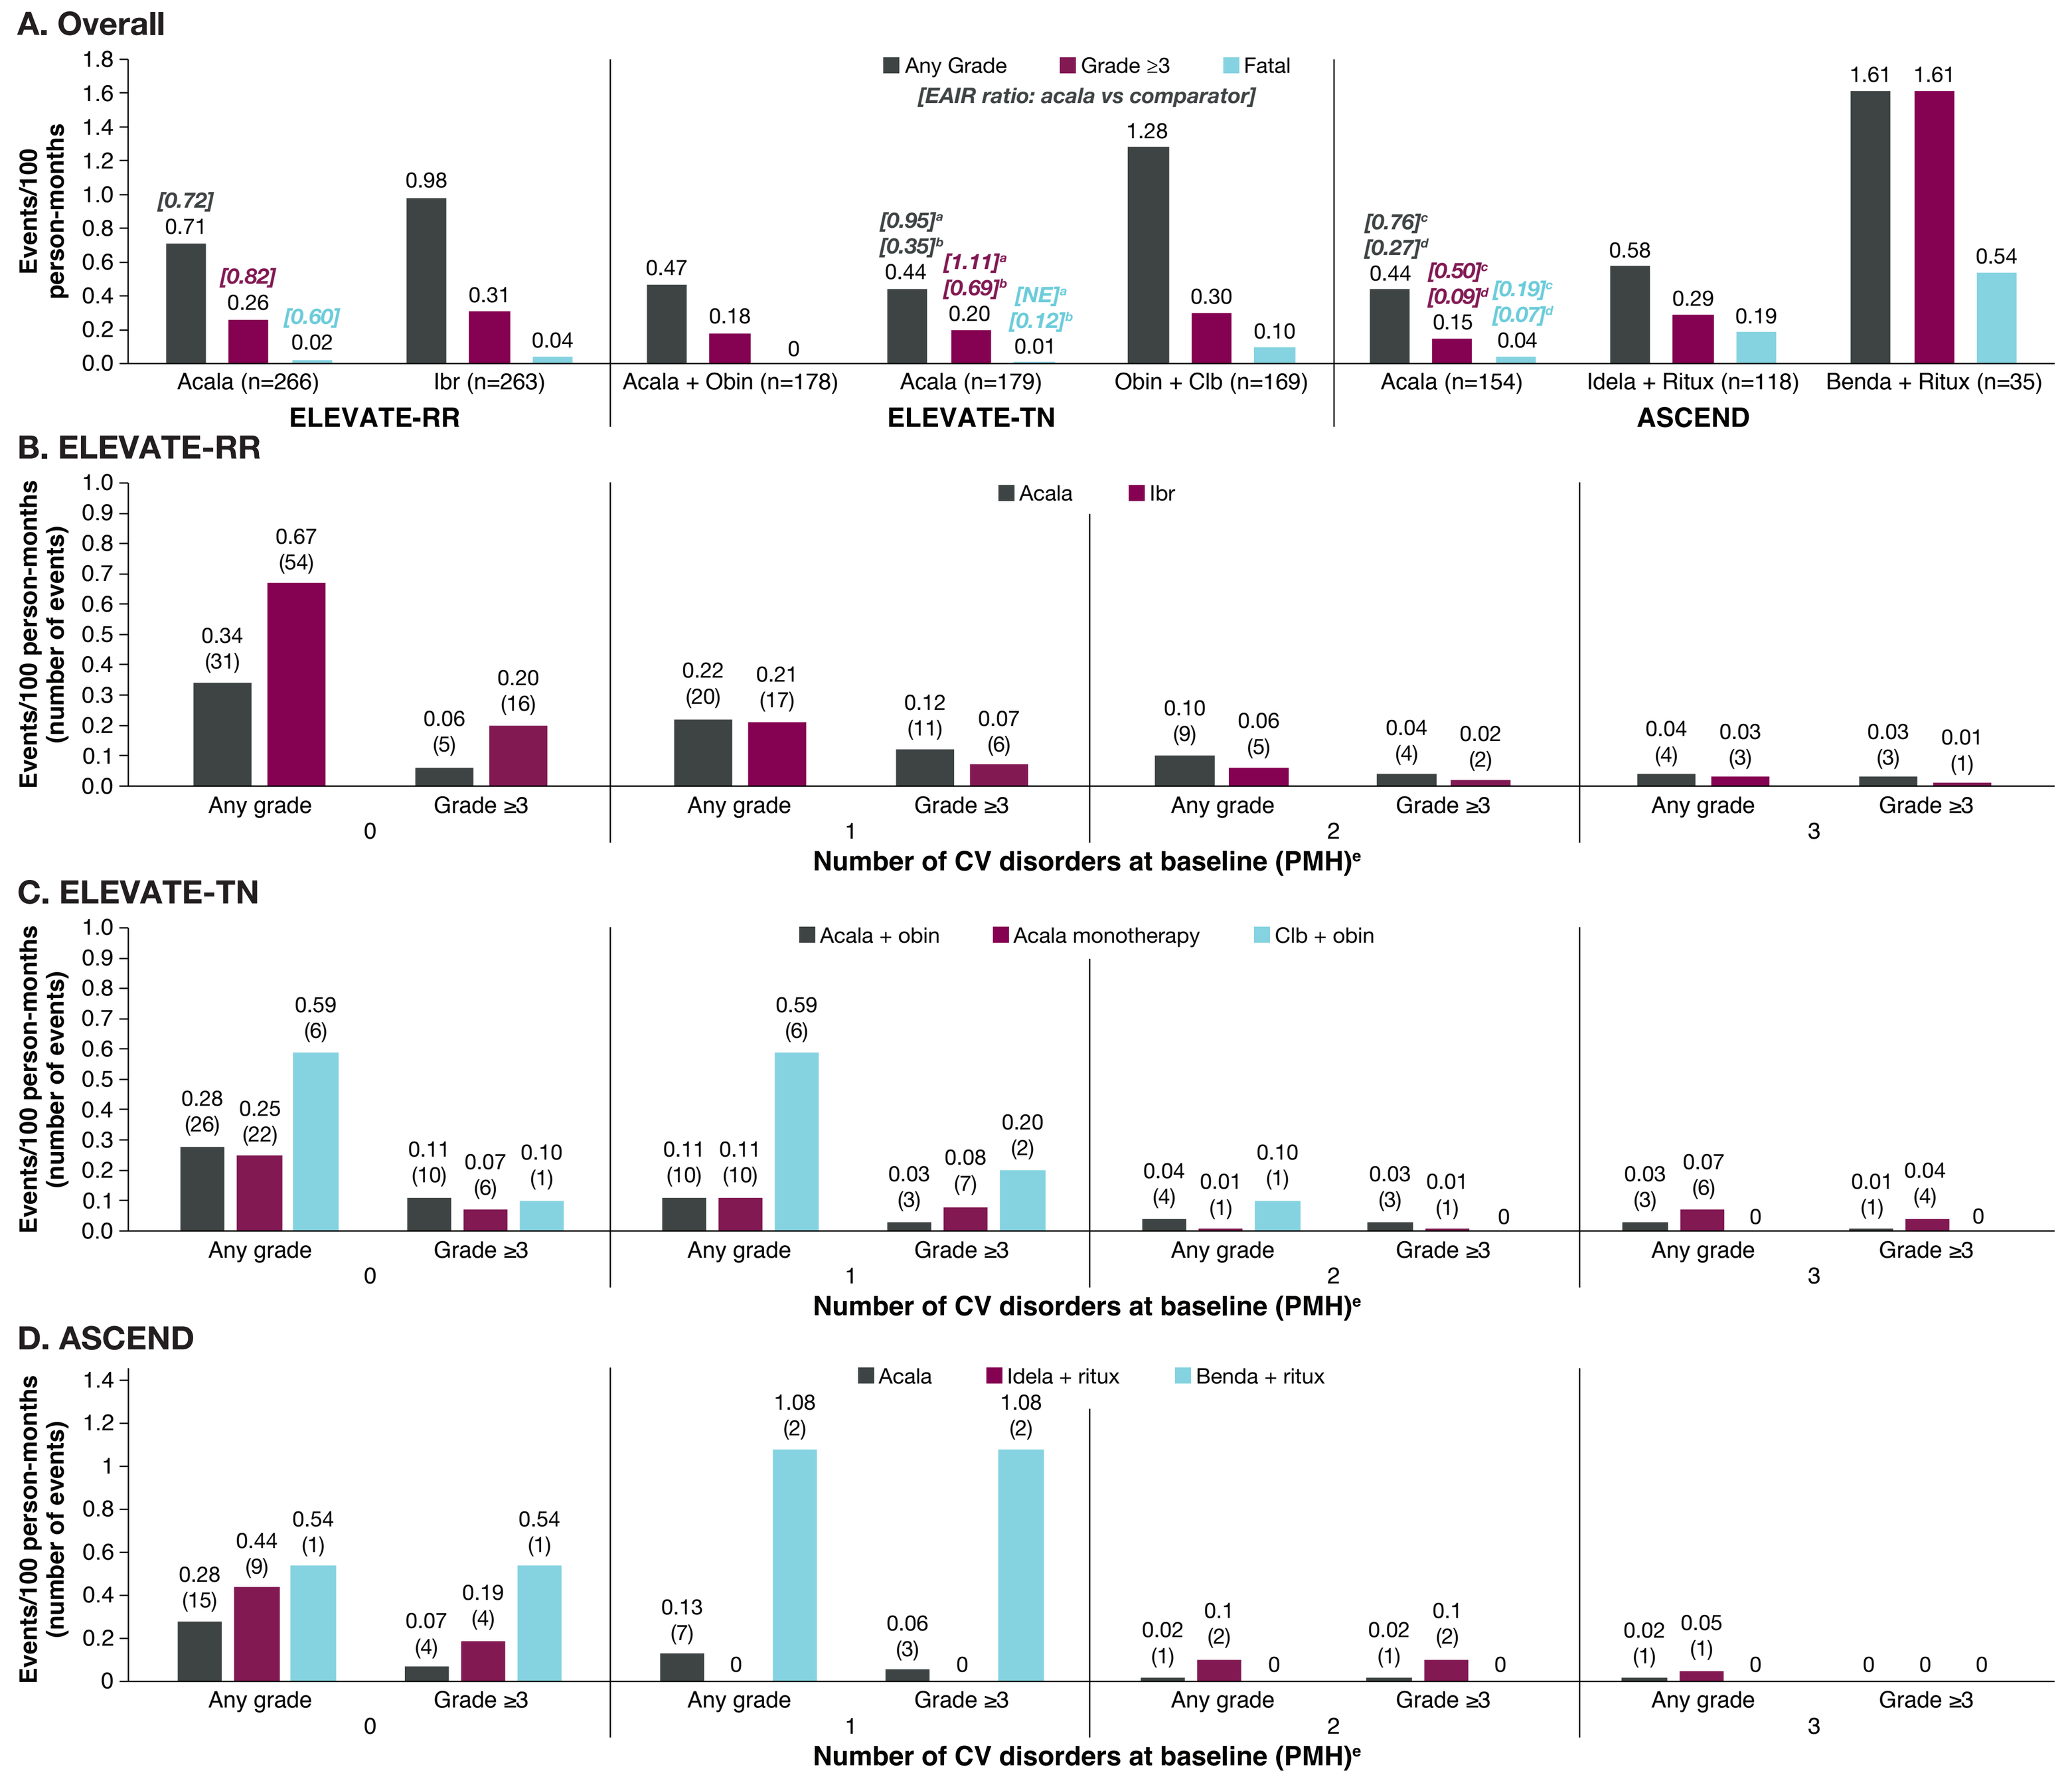

A. Overall
Any Grade
Grade ≥3
Fatal
[EAIR ratio: acala vs comparator]
Events/100 person-months
Acala (n=266)
Ibr (n=263)
ELEVATE-RR
Acala + Obin (n=178)
Acala (n=179)
Obin + Clb (n=169)
ELEVATE-TN
Acala (n=154)
Idela + Ritux (n=118)
Benda + Ritux (n=35)
ASCEND
B. ELEVATE-RR
Acala
Ibr
C. ELEVATE-TN
Acala + obin
Acala monotherapy
Clb + obin
D. ASCEND
Acala
Idela + ritux
Benda + ritux
Events/100 person-months (number of events)
Any grade
Grade ≥3
Number of CV disorders at baseline (PMH)[e]

Each patient/event is counted only once under maximum grade. [a]Ratio of acala monotherapy to acala + obin. [b]Ratio of acala monotherapy to obin + clb. [c]Ratio of acala monotherapy to idela + ritux. [d]Ratio of acala monotherapy to benda + ritux. [e]Number of CV disorders at baseline is based on CV PTs from PMH, as described in **Supplemental Table 2**.

Table 2. Most frequent (EAIR ≥0.03) treatment-emergent cardiac disorder events (PT) in pooled acalabrutinib and pooled comparator groups

Cardiac disorder PT	Acalabrutinib pooled (n=599) Any grade	Acalabrutinib pooled (n=599) Grade ≥3	Acalabrutinib pooled (n=599) Fatal	Comparator pooled (n=585) Any grade	Comparator pooled (n=585) Grade ≥3	Comparator pooled (n=585) Fatal
Atrial fibrillation	47 (0.20)	16 (0.07)	0	46 (0.41)	10 (0.09)	0
Palpitations	19 (0.08)	0	0	13 (0.12)	0	0
Cardiac failure	10 (0.04)	7 (0.03)	0	9 (0.08)	7 (0.06)	1 (0.01)
Tachycardia	10 (0.04)	0	0	9 (0.08)	0	0
Angina pectoris	13 (0.06)	4 (0.02)	0	6 (0.05)	2 (0.02)	0
Sinus tachycardia	2 (0.01)	0	0	6 (0.05)	0	0
Cardiac failure chronic	2 (0.01)	1 (0.00)	0	5 (0.04)	3 (0.03)	1 (0.01)
Myocardial ischemia	3 (0.01)	1 (0.00)	0	4 (0.04)	2 (0.02)	0
Acute myocardial infarction	3 (0.01)	3 (0.01)	0	3 (0.03)	2 (0.02)	0
Arrhythmia	4 (0.02)	1 (0.00)	0	3 (0.03)	0	0
Atrial flutter	3 (0.01)	1 (0.00)	0	3 (0.03)	2 (0.02)	0
Cardiac arrest	1 (0.00)	1 (0.00)	0	3 (0.03)	3 (0.03)	3 (0.03)
Coronary artery disease	1 (0.00)	1 (0.00)	0	3 (0.03)	2 (0.02)	0
Mitral valve incompetence	0	0	0	3 (0.03)	2 (0.02)	0
Myocardial infarction	3 (0.01)	3 (0.01)	1 (0.00)	3 (0.03)	3 (0.03)	1 (0.01)
Pericarditis	1 (0.00)	0	0	3 (0.03)	2 (0.02)	0
Sinus bradycardia	6 (0.03)	0	0	3 (0.03)	0	0

Number of events (EAIR). Each patient/event is counted only once under maximum grade.

Figure 3. EAIR of treatment-emergent cardiac disorder events (SOC) for pooled acalabrutinib and pooled comparator groups

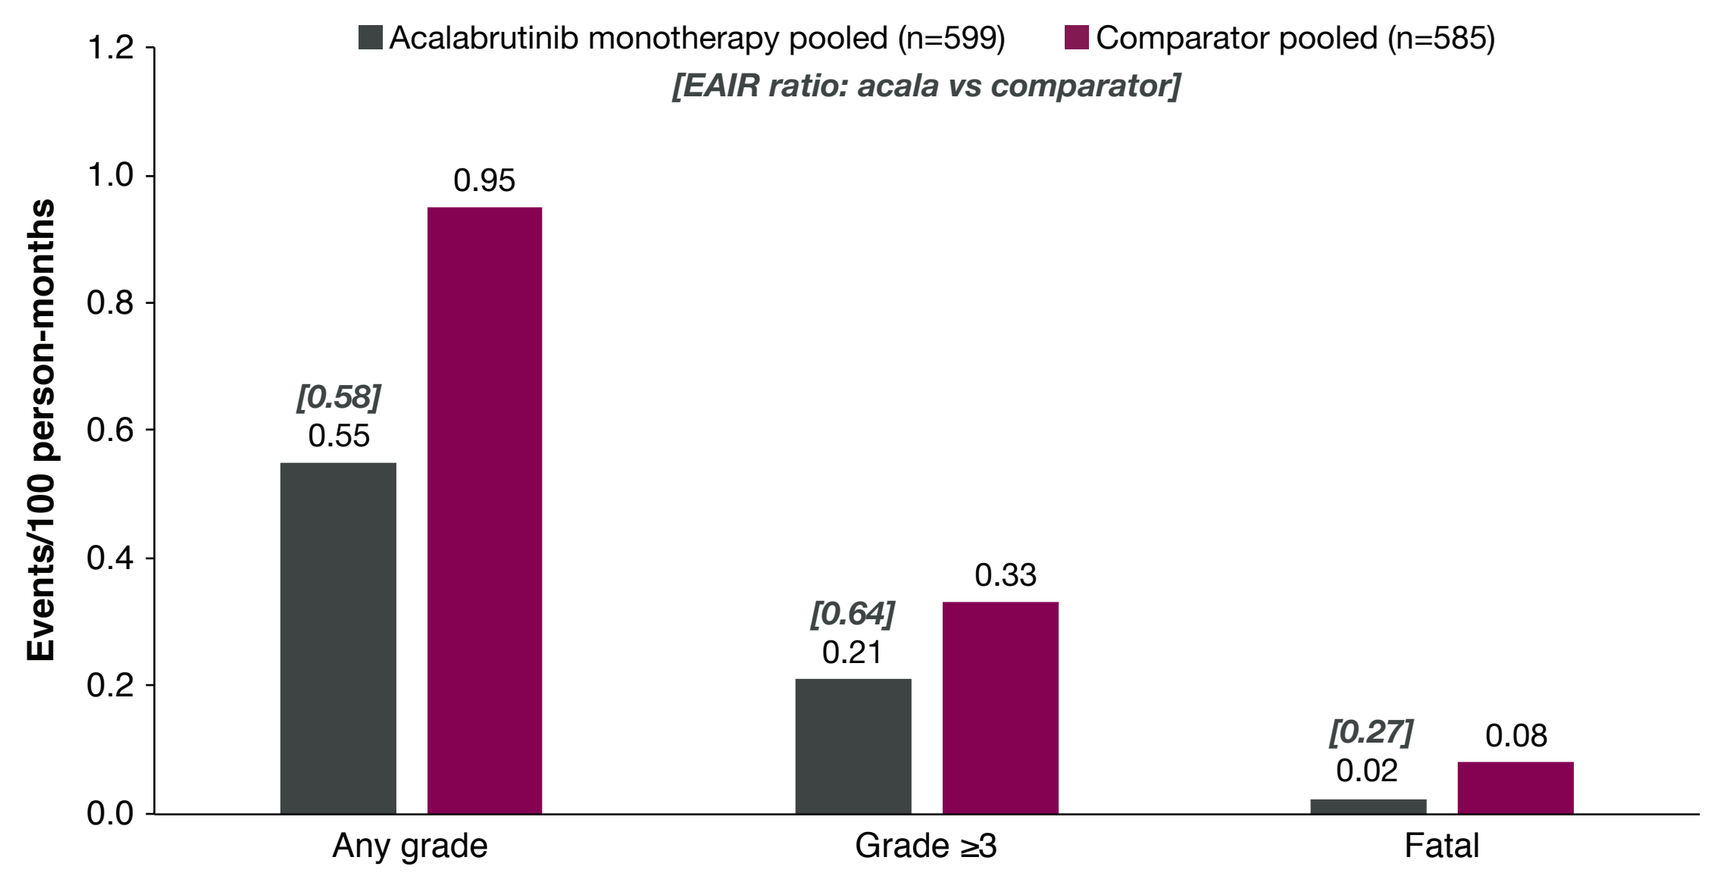

Acalabrutinib monotherapy pooled (n=599)
Comparator pooled (n=585)
[EAIR ratio: acala vs comparator]
Events/100 person-months
[0.58]
0.55
0.95
[0.64]
0.21
0.33
[0.27]
0.02
0.08
Any grade
Grade ≥3
Fatal

Figure 4. EAIR of cardiac disorder events (SOC) for patients who crossed over to acalabrutinib from comparator (ELEVATE-TN and ASCEND)

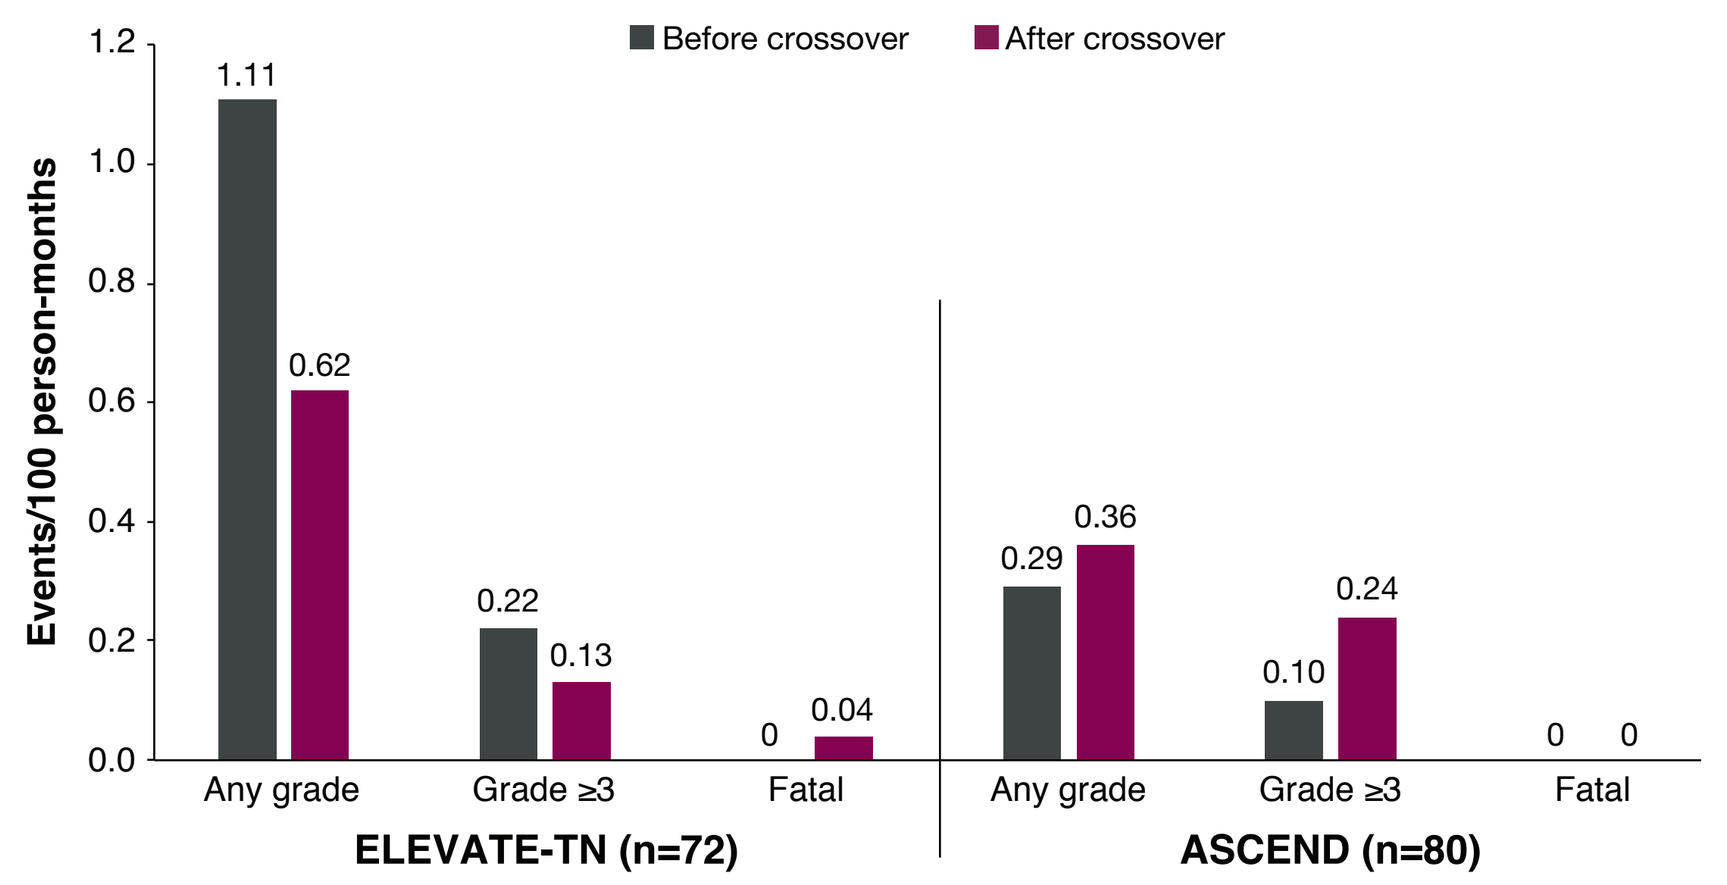

Before crossover
After crossover
Events/100 person-months
1.11
0.62
0.22
0.13
0
0.04
0.29
0.36
0.10
0.24
0
0
Any grade
Grade ≥3
Fatal
ELEVATE-TN (n=72)
ASCEND (n=80)

Table 3. EAIR of treatment-emergent cardiac disorder events (SOC) for pooled acalabrutinib and pooled comparator groups by number of CV disorders at baseline

Number of CV disorders at baseline (PMH)[b]	Acalabrutinib pooled (n=599) Any grade	Acalabrutinib pooled (n=599) Grade ≥3	Acalabrutinib pooled (n=599) Fatal	Comparator pooled (n=585) Any grade	Comparator pooled (n=585) Grade ≥3	Comparator pooled (n=585) Fatal
0	68 (0.29)	15 (0.06)	4 (0.02)	70 (0.62)	22 (0.19)	7 (0.06)
1	37 (0.16)	21 (0.09)	0	25 (0.22)	10 (0.09)	1 (0.01)
2	11 (0.05)	6 (0.03)	1 (0.00)	8 (0.07)	4 (0.04)	1 (0.01)
≥3	11 (0.05)	7 (0.03)	0	4 (0.04)	1 (0.01)	0

Number of events (EAIR)[a]. Each patient/event is counted only once under maximum grade.
[a]Based on cardiac TEAEs under SOC "cardiac disorders."
[b]Number of CV disorders at baseline is based on CV PTs from PMH, as described in **Supplemental Table 2**.

Table 4. EAIR of treatment-emergent cardiac disorder events (SOC) for patients who crossed over to acalabrutinib from comparator by number of CV disorders at baseline (ELEVATE-TN and ASCEND)

Number of CV disorders at baseline (PMH)[b]	ELEVATE-TN Before crossover (n=72) Any grade	Grade ≥3	Fatal	ELEVATE-TN After crossover (n=72) Any grade	Grade ≥3	Fatal	ASCEND Before crossover (n=80) Any grade	Grade ≥3	Fatal	ASCEND After crossover (n=80) Any grade	Grade ≥3	Fatal
0	2 (0.44)	0	0	9 (0.40)	1 (0.04)	0	2 (0.19)	1 (0.10)	0	3 (0.18)	2 (0.12)	0
1	3 (0.67)	1 (0.22)	0	3 (0.13)	1 (0.04)	0	0	0	0	1 (0.06)	1 (0.06)	0
2	0	0	0	2 (0.09)	1 (0.04)	1 (0.04)	0	0	0	1 (0.06)	1 (0.06)	0
≥3	0	0	0	0	0	0	1 (0.10)	0	0	1 (0.06)	0	0

Number of events (EAIR)[a]. Each patient/event is counted only once under maximum grade.
[a]Based on cardiac TEAEs under SOC "cardiac disorders."
[b]Number of CV disorders at baseline is based on CV PTs from PMH, as described in **Supplemental Table 2**.

Abbreviations

Acala, acalabrutinib; Benda, bendamustine; BMI, body mass index; BTK, Bruton tyrosine kinase; BTKi, Bruton tyrosine kinase inhibitor; Clb, chlorambucil; CLL, chronic lymphocytic leukemia; CV, cardiovascular; EAIR, exposure-adjusted incidence rate; Ibr, ibrutinib; Idela, idelalisib; N/A, not applicable; NE, not evaluable; Obin, obinutuzumab; PMH, past medical history; PT, preferred term; Ritux, rituximab; RCT, randomized controlled trial; R/R, relapsed/refractory; SMQ, Standardized Medical Dictionary for Regulatory Activities Query; SOC, system organ class; TEAE, treatment-emergent adverse event; TN, treatment-naive.

Acknowledgments

This analysis was sponsored by AstraZeneca. Medical writing and editorial support was provided by Robert J Schoen, PharmD, and Cindy Gobbel, PhD, of Peloton Advantage, LLC, an OPEN Health company, and funded by AstraZeneca. The authors directed development of the presentation and are fully responsible for all content and editorial decisions.

References

1. Broccoli A, et al. *J Clin Med.* 2021;10:5845. 2. Imbruvica [package insert]. South San Francisco, CA: Pharmacyclics LLC; August 2022. 3. Calquence [package insert]. Wilmington, DE: AstraZeneca Pharmaceuticals; August 2022. 4. Byrd JC, et al. *J Clin Oncol.* 2021;39:3441-52. 5. Lipsky A, Lamanna N. *Hematology Am Soc Hematol Educ Program.* 2020;2020:336-45. 6. Sharman JP, et al. *Lancet.* 2020;395:1278-91. 7. Ghia P, et al. *J Clin Oncol.* 2020;38:2849-61.